An Allosteric, Orally Administered CBL-B Inhibitor Remodels the Tumor Microenvironment and Enhances Immune-mediated Tumor Growth Inhibition

#424

Yilin Qi*, Jun Kuai*, Yingzhi Bi*, Huadong Sun, Samira Jaeger, David Greco, Ken Carson, Timothy Reilly, Geraldine Harriman, Fang Wang†

HotSpot Therapeutics, Inc, 50 Milk St, 16th floor, Boston, MA 02109, USA; *equal contribution; †corresponding author

Introduction

- Casitas B-lineage lymphoma proto-oncogene b (CBL-B), an E3 ubiquitin-protein ligase, is a critical regulator of immunity.
- Genetic ablation or inactivation of CBL-B bypasses the requirement of a co-stimulatory signal for T cell activation in an antigen-dependent manner. As a consequence, *CBLB* knockout mice spontaneously reject tumor growth, and the effect is largely dependent on CD8+T cells. Depletion of *CBLB* gene in human CD8+T cells augments the antigen-specific tumor killing.
- The extensive *in vitro* and *in vivo* evidence suggest inhibition of CBL-B may present an exciting opportunity to enhance immune-mediated tumor suppression.
- HotSpot has identified a series of allosteric CBL-B inhibitors which exhibited potent *in vitro* and *in vivo* properties.

CBL-B Inhibition Enhances Anti-Tumor Immunity Through Several Key Biological Mechanisms

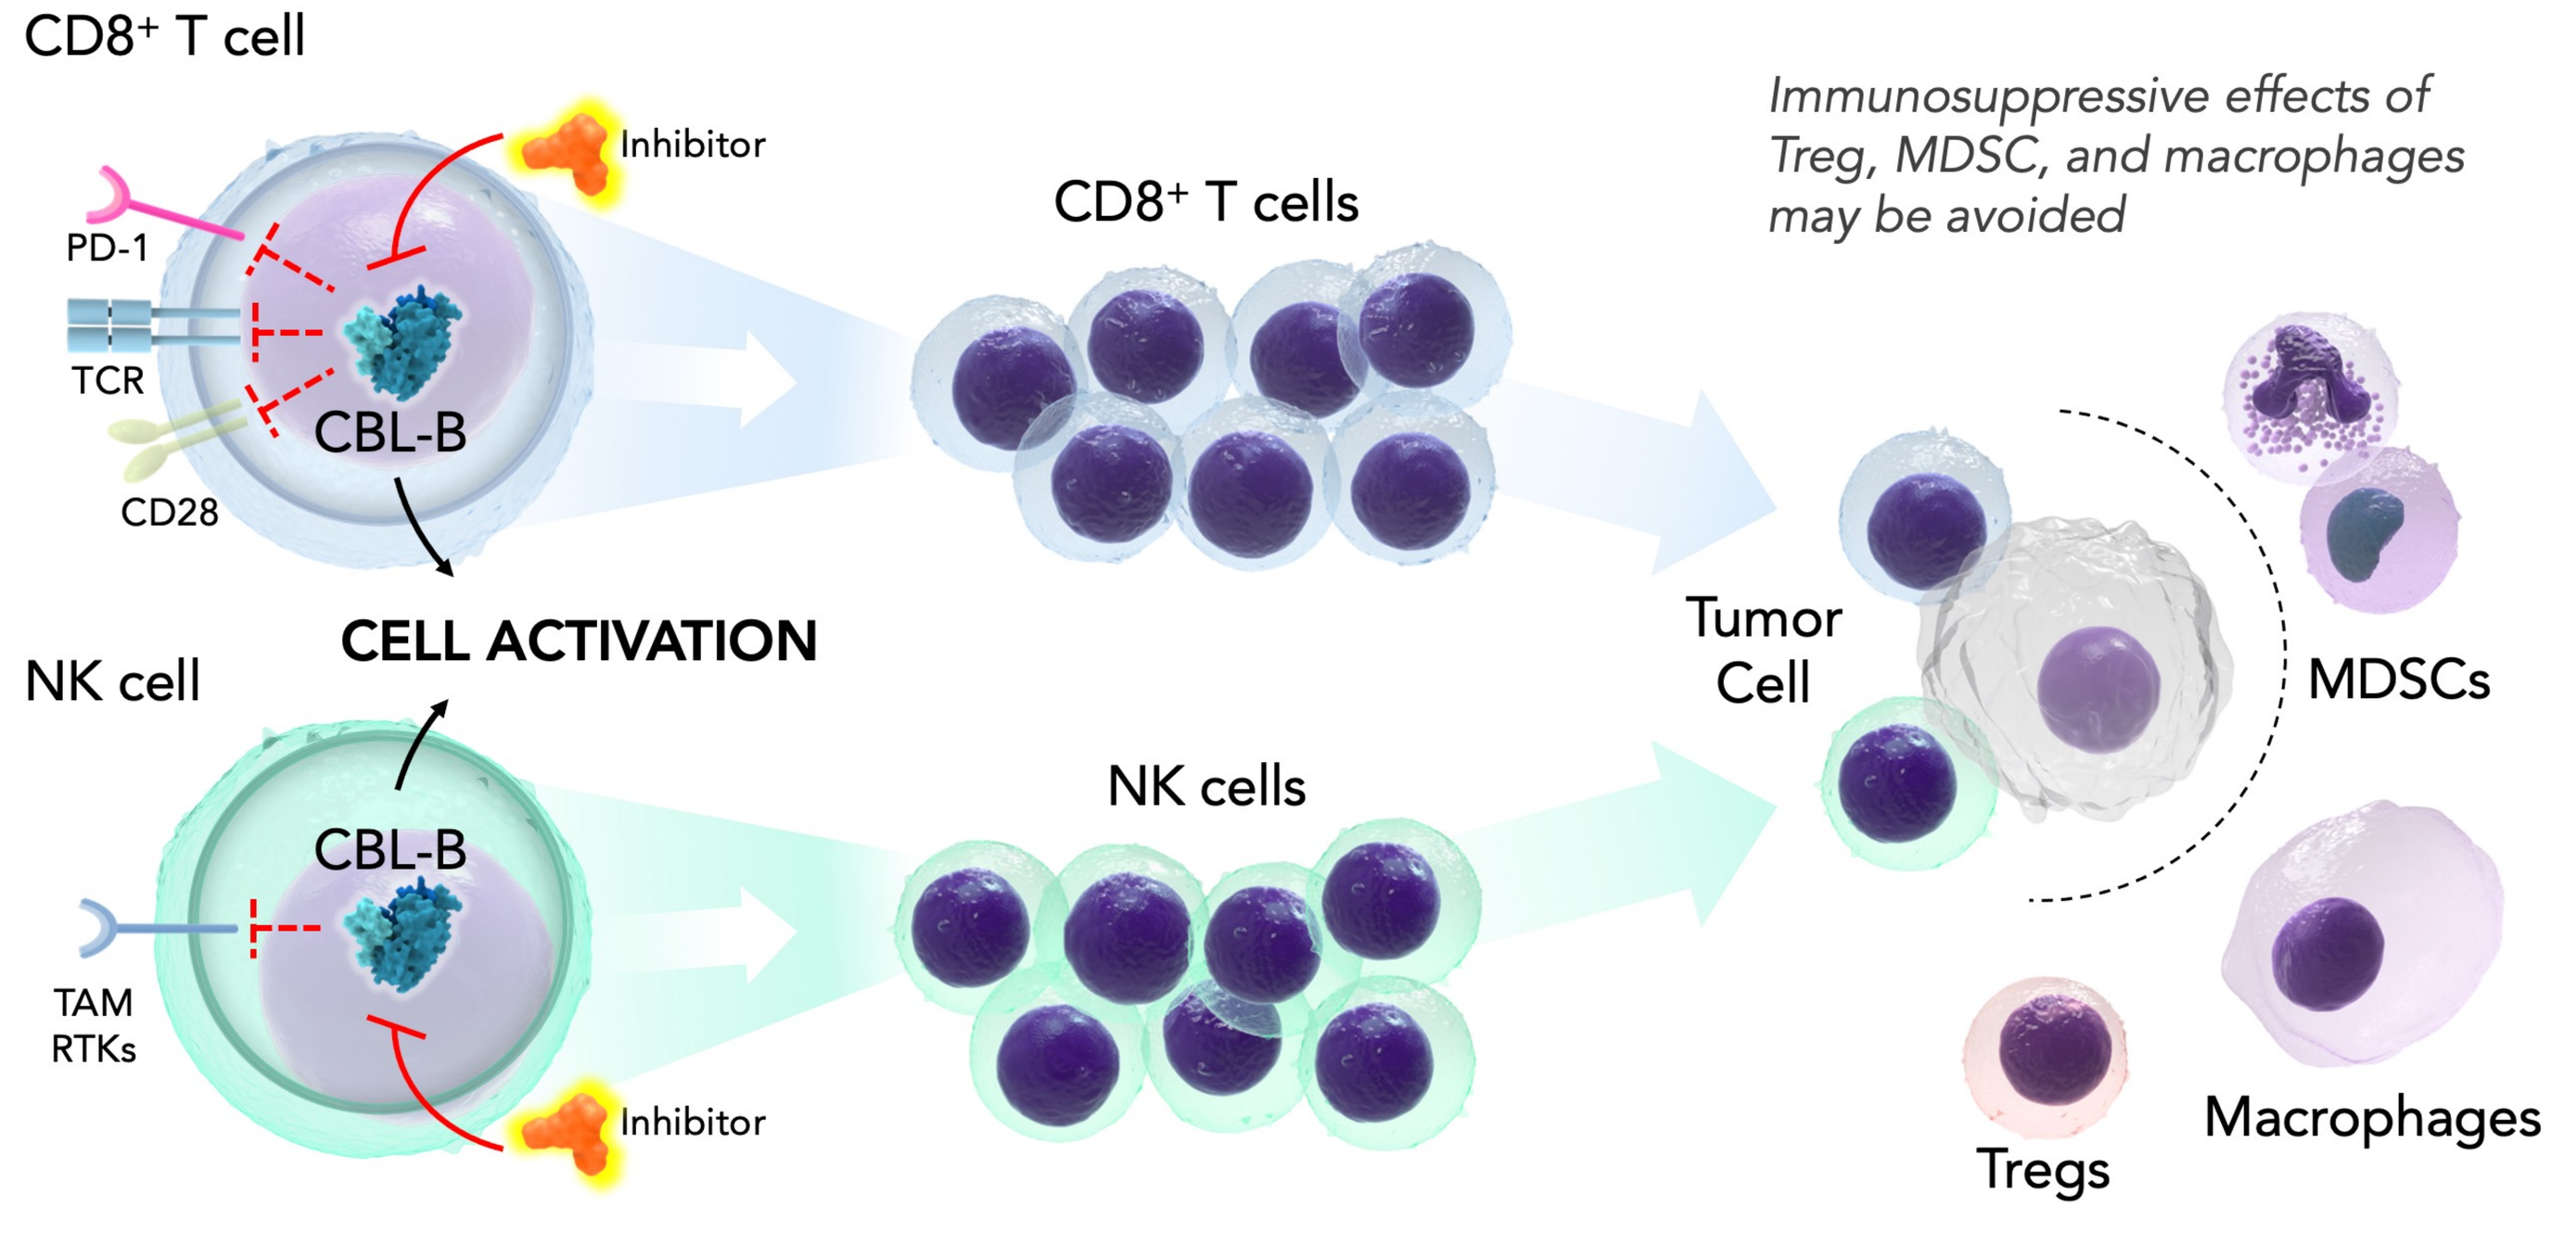
CD8⁺ T cell
Inhibitor
PD-1
TCR
CBL-B
CD28
CELL ACTIVATION
NK cell
CBL-B
TAM RTKs
Inhibitor
CD8⁺ T cells
NK cells
Immunosuppressive effects of Treg, MDSC, and macrophages may be avoided
Tumor Cell
MDSCs
Macrophages
Tregs

1 Lowers threshold for effector cell activation

2 Drives effector cell proliferation and inflammation

3 Reduces susceptibility to suppression

Method

- HotSpot's allosteric CBL-B inhibitor HOT-A was evaluated in a set of syngeneic mouse tumor models
 - H22 murine liver cancer model in female BALB/c mice (H22)
 - CT26 murine colorectal cancer model in female BALB/c mice (CT26)
 - B16F10 murine melanoma cancer model in female C57BL/6 mouse (B16F10)
 - LL/2 murine lung carcinoma model in female C57BL/6 mouse (LL2)
- Tumor growth was compared between vehicle treated animals and animals treated with HOT-A.
- Tumor gene expression was characterized by nanostring analysis.
- Immunohistochemistry and flow-based immunophenotyping were used in profiling the tumor microenvironment.

Results

Figure 1. HotSpot CBL-B Inhibitor, HOT-A, Showed Single Agent Efficacy in Multiple Tumor Models

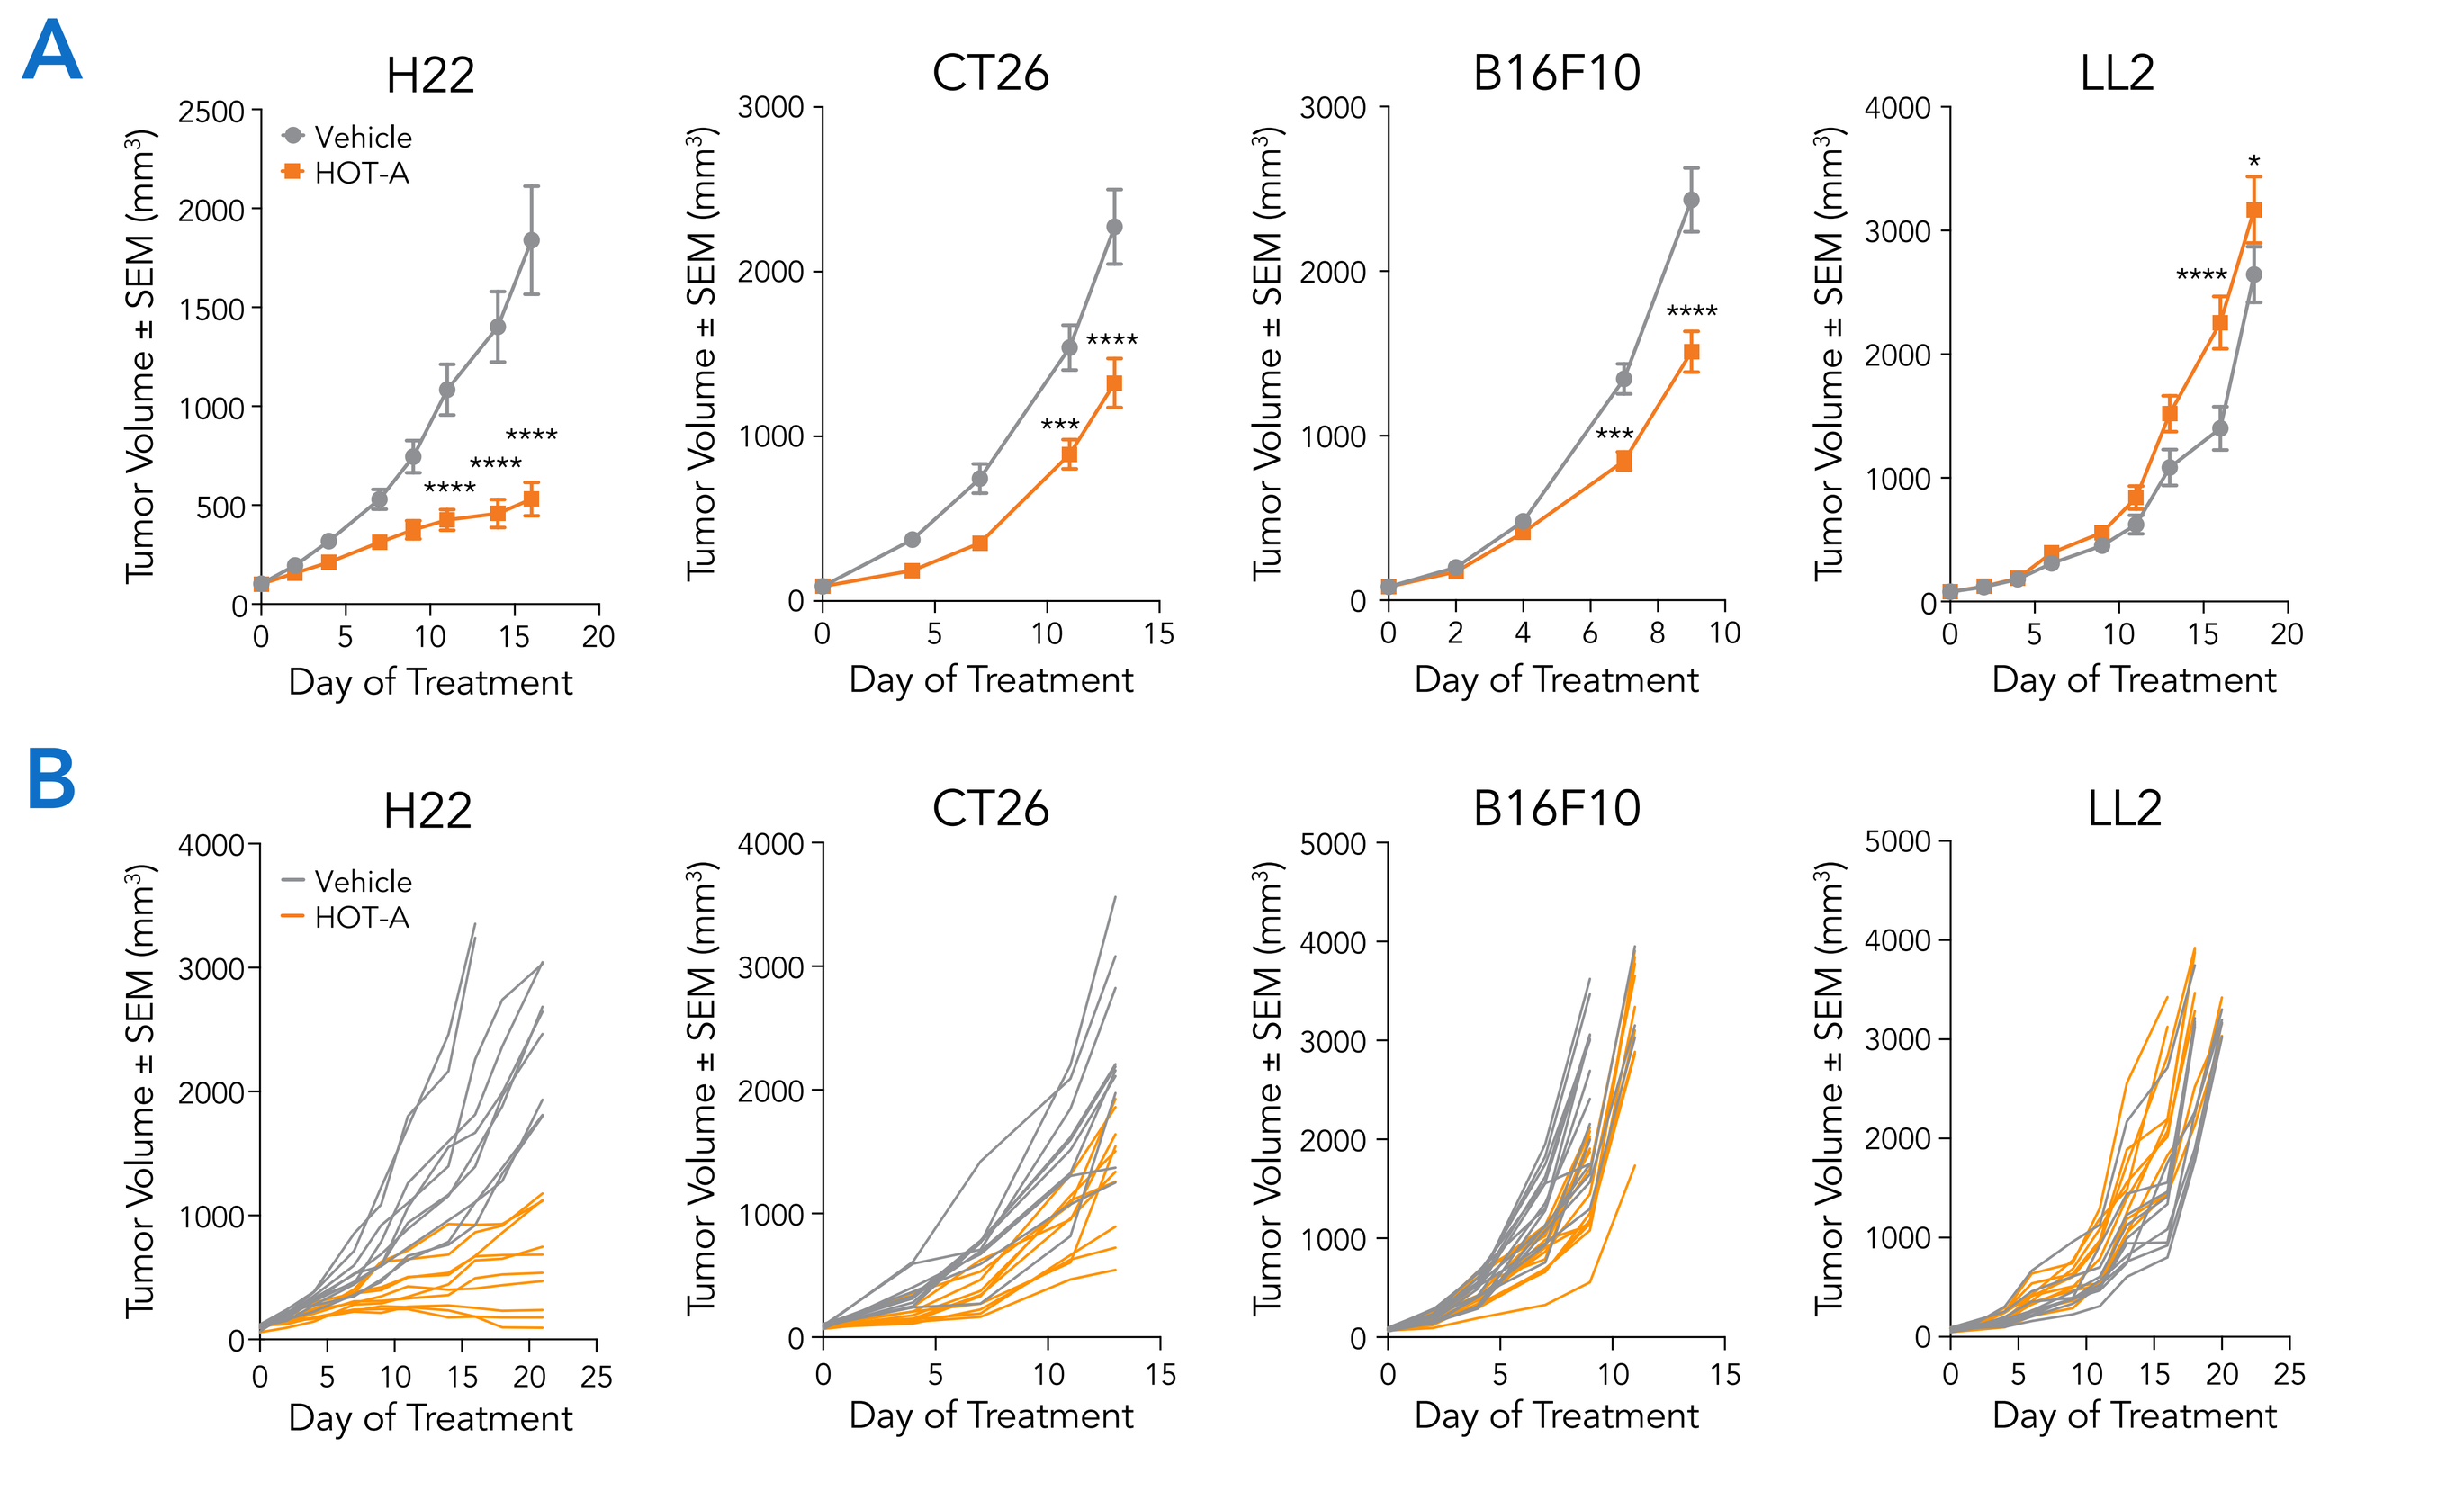
A
H22
CT26
B16F10
LL2
Vehicle
HOT-A
Tumor Volume ± SEM (mm³)
Day of Treatment
B
H22
CT26
B16F10
LL2
Vehicle
HOT-A
Tumor Volume ± SEM (mm³)
Day of Treatment

Female BALB/c (H22 and CT26 tumor model) or C57BL/6 (B16F10 and LL2 tumor model) mice were inoculated with tumor cells, randomized, and treated with vehicle or HOT-A. (A) Tumor growth of vehicle or HOT-A treated animals. Data are displayed as mean ± standard error of the mean (SEM). Statistics were calculated using 2-way ANOVA. Only statistically significant differences among vehicle versus HOT-A treated groups are shown. *$p<0.05$; ***$p<0.0001$; ****$p<0.00001$. (B) Individual tumor growth curves for vehicle and treated groups in each tumor model.

Figure 2. HOT-A Responding Tumor Models Showed Increased Inflammatory Gene Signature

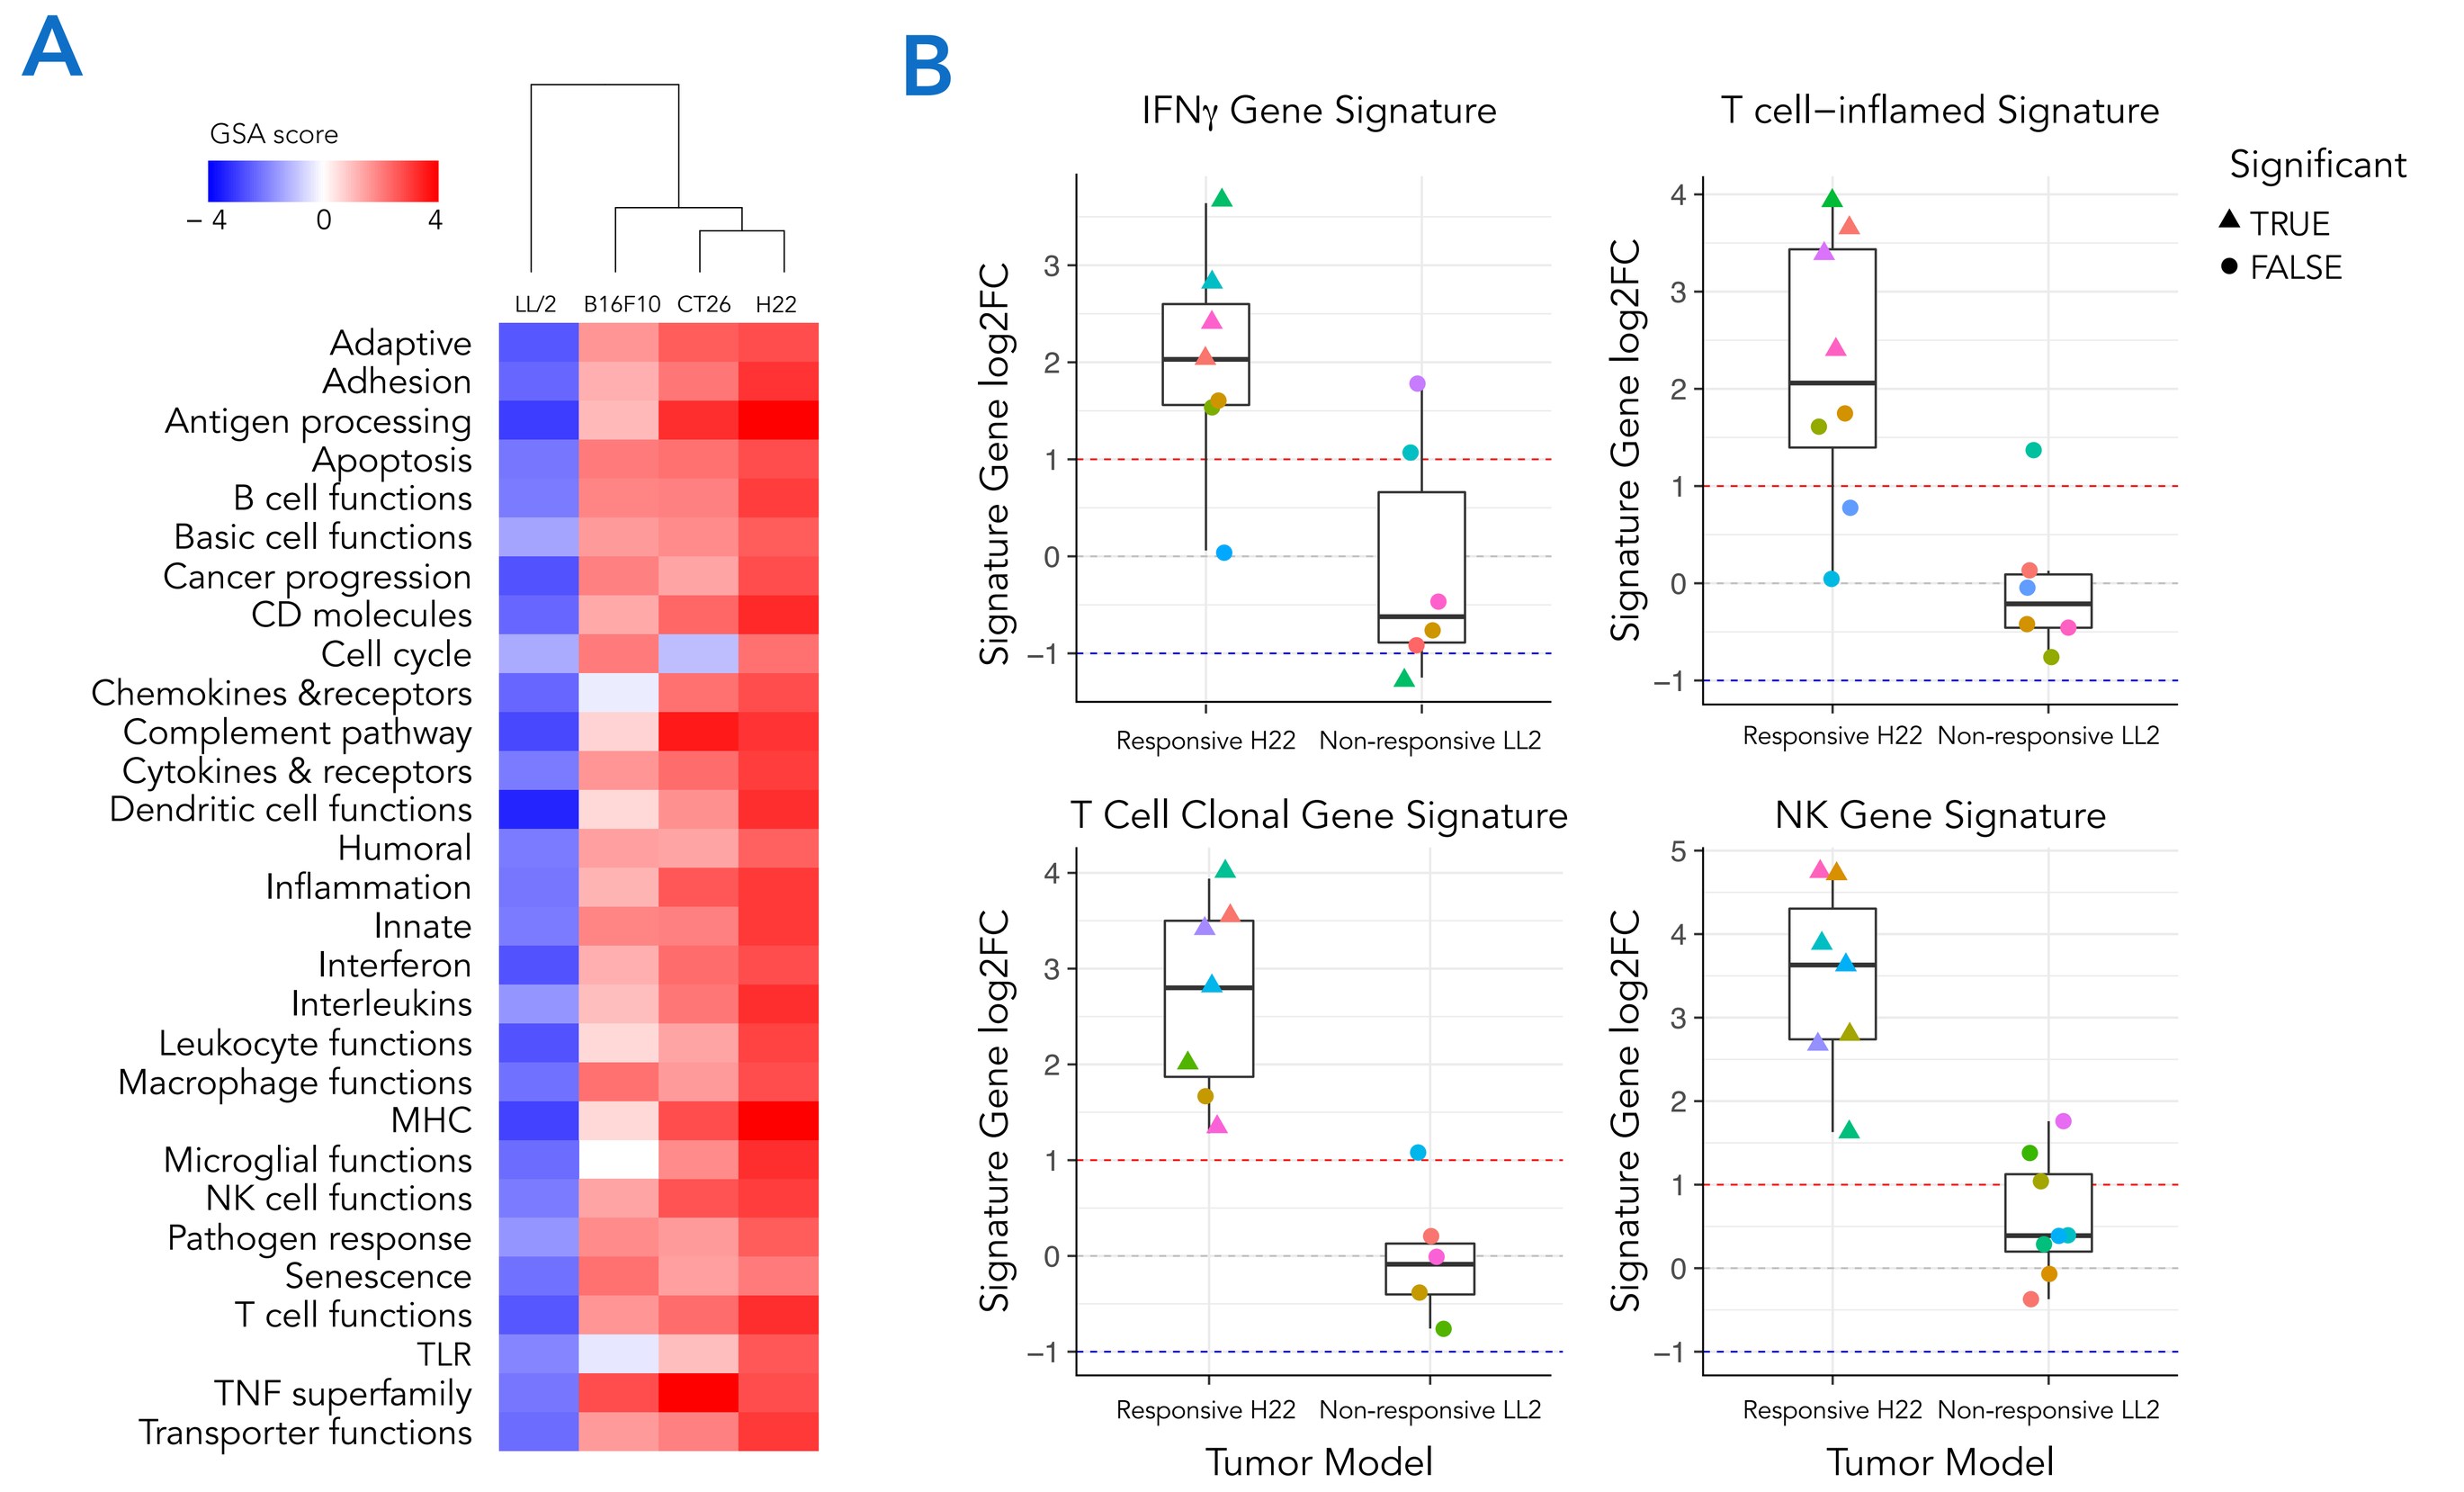
A
GSA score
LL/2
B16F10
CT26
H22
Adaptive
Adhesion
Antigen processing
Apoptosis
B cell functions
Basic cell functions
Cancer progression
CD molecules
Cell cycle
Chemokines &receptors
Complement pathway
Cytokines & receptors
Dendritic cell functions
Humoral
Inflammation
Innate
Interferon
Interleukins
Leukocyte functions
Macrophage functions
MHC
Microglial functions
NK cell functions
Pathogen response
Senescence
T cell functions
TLR
TNF superfamily
Transporter functions
B
IFNγ Gene Signature
T cell–inflamed Signature
T Cell Clonal Gene Signature
NK Gene Signature
Significant
TRUE
FALSE
Signature Gene log2FC
Responsive H22
Non-responsive LL2
Tumor Model

At the end of tumor efficacy study (Figure 1), tumor mRNA was extracted and analyzed using nanoString™ mouse PanCancer immune profiling panel. (A) Immune response signature scores based on the differential gene expression analysis between HOT-A treated tumor vs vehicle treated tumors. (B) Comparison of log2 fold change (log2FC) of IFNγ, T cell activation, T cell clonal expansion and NK function signature genes between responsive H22 and non-responsive LL2 tumors

Figure 3. HOT-A Treated H22 Tumor Showed Increased Immune Cell Infiltration

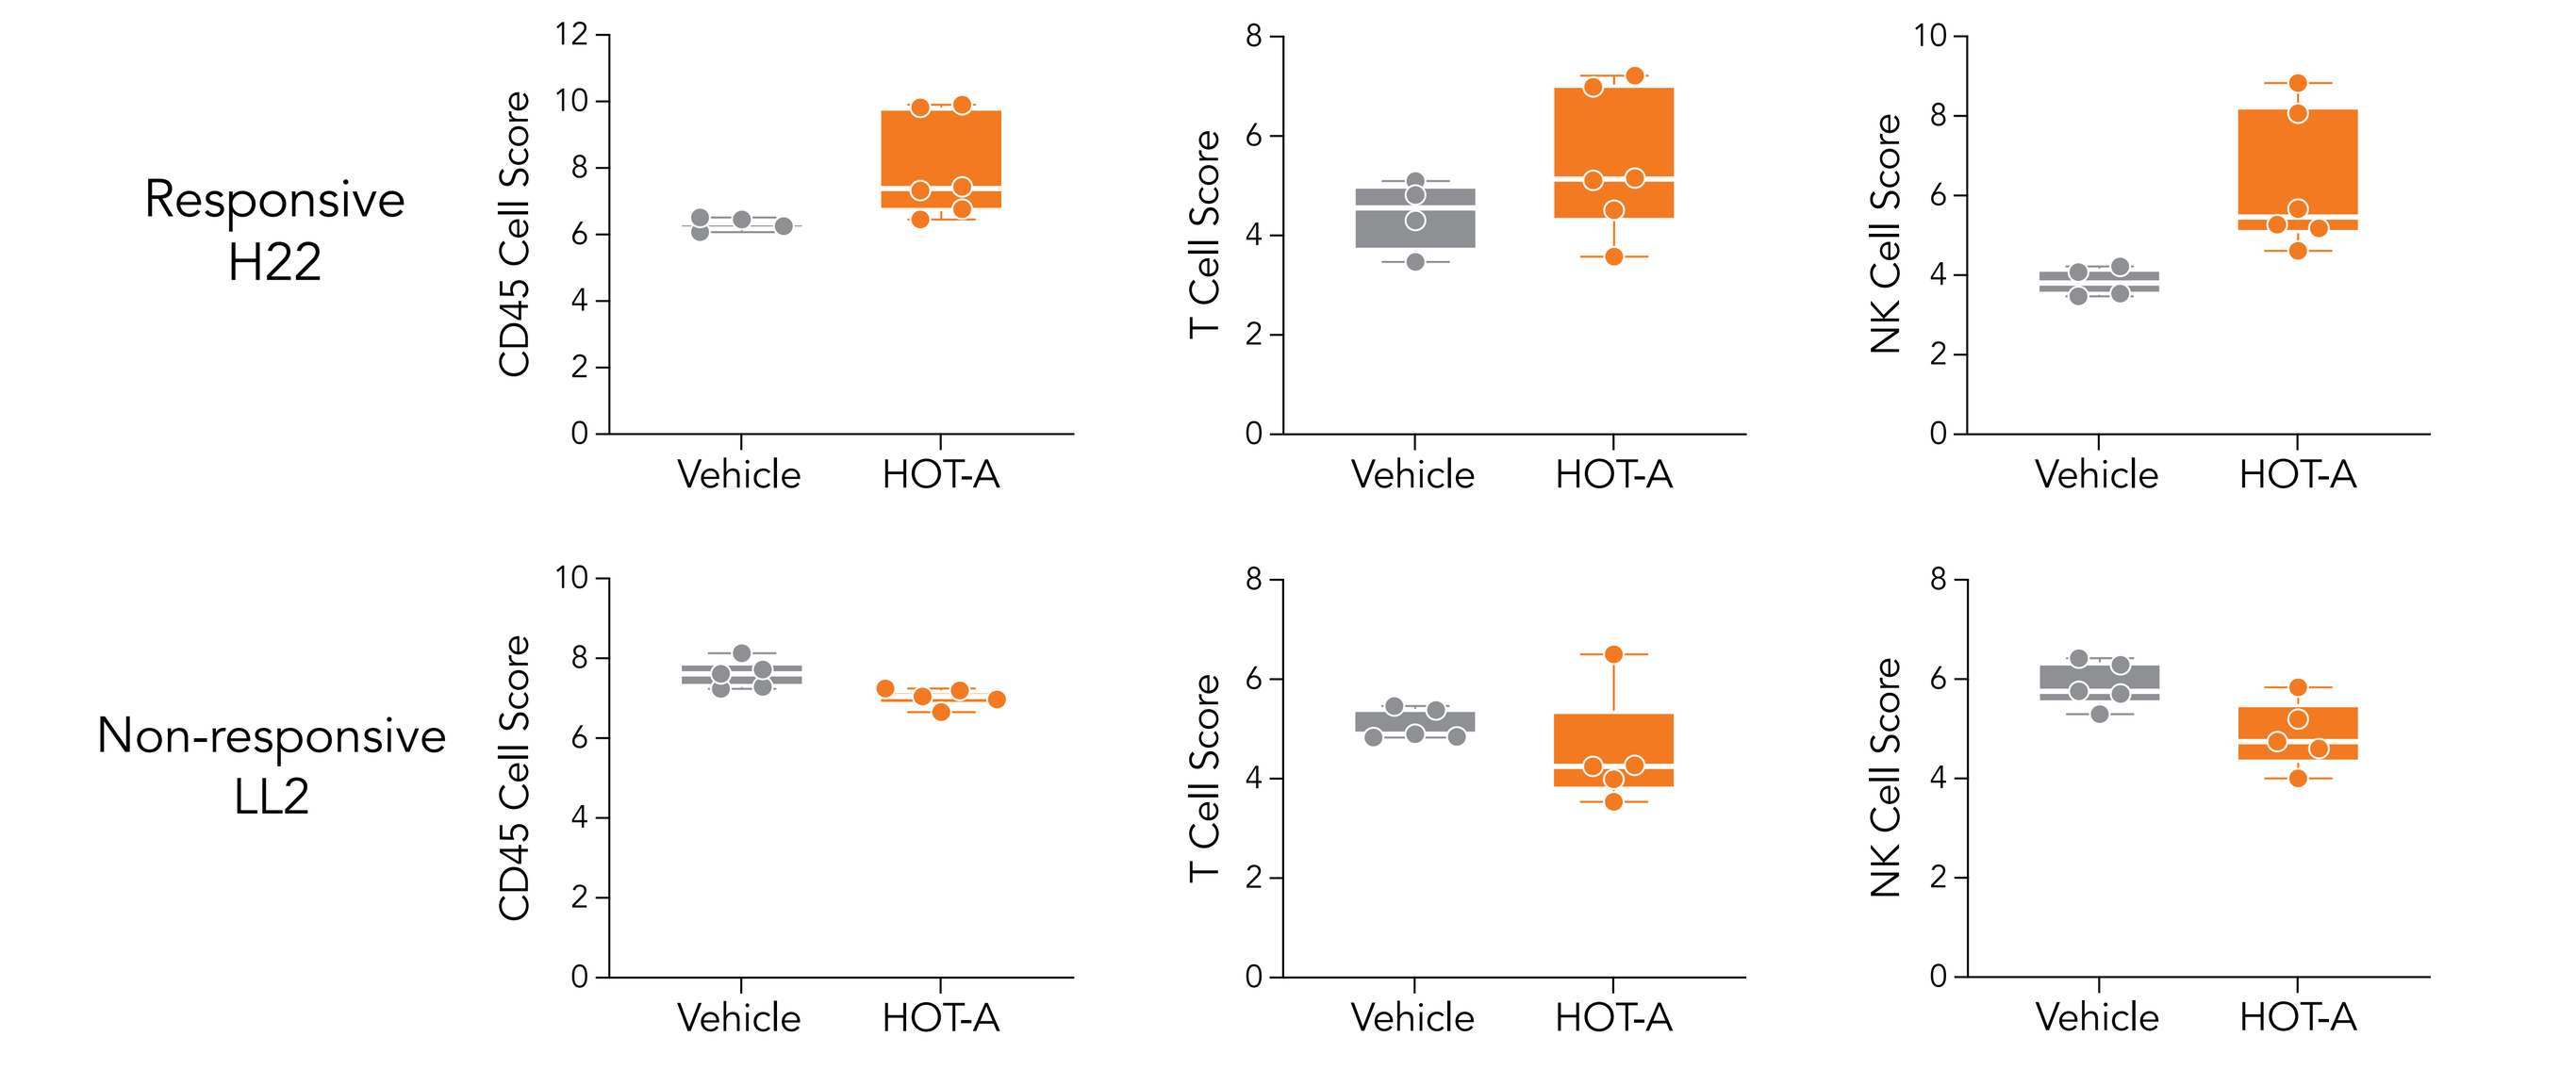
Responsive H22
Non-responsive LL2
CD45 Cell Score
T Cell Score
NK Cell Score
Vehicle
HOT-A

At the end of H22 and LL2 tumor efficacy study, tumor mRNA was extracted and analyzed using nanoString™ mouse PanCancer immune profiling panel. CD45, T cell and NK cell scores were derived from the nanostring tumor analysis.

Figure 4. HOT-A Mediated Tumor Inhibition Requires Competent Immune System

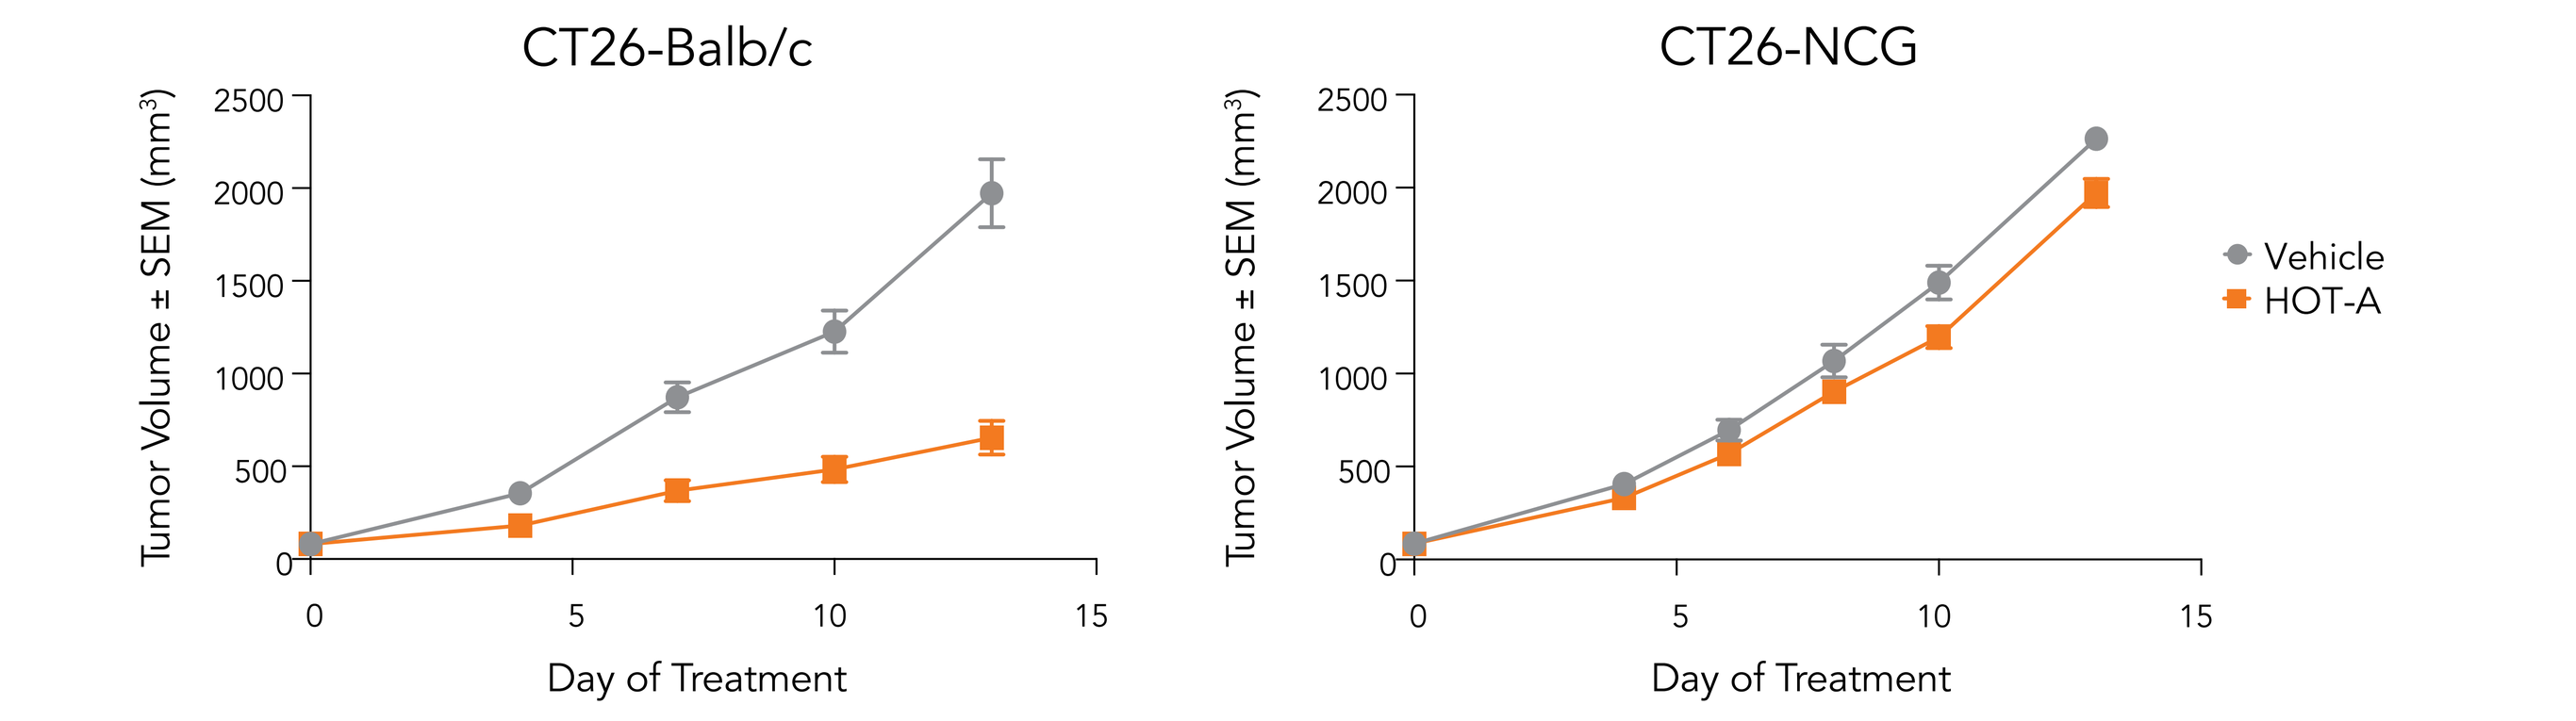
CT26-Balb/c
CT26-NCG
Tumor Volume ± SEM (mm³)
Day of Treatment
Vehicle
HOT-A

Female BALB/c mice or NCG mice (NOD-*Prkdc*^{em26Cd52}*Il2rg*^{em26Cd22}/NjuCrl, coisogenic immunodeficient, lack functional/mature T, B, and NK cells, and have reduced macrophage and dendritic cell function) were inoculated with CT26 tumor cells, randomized, and treated with vehicle or HOT-A. Tumor growth curves for vehicle treated animals and animals treated with HOT-A are shown. Data are displayed as mean ± standard error the mean (SEM).

Figure 5. Combination of HOT-A and Anti-PD1 Further Enhanced Tumor Inhibition

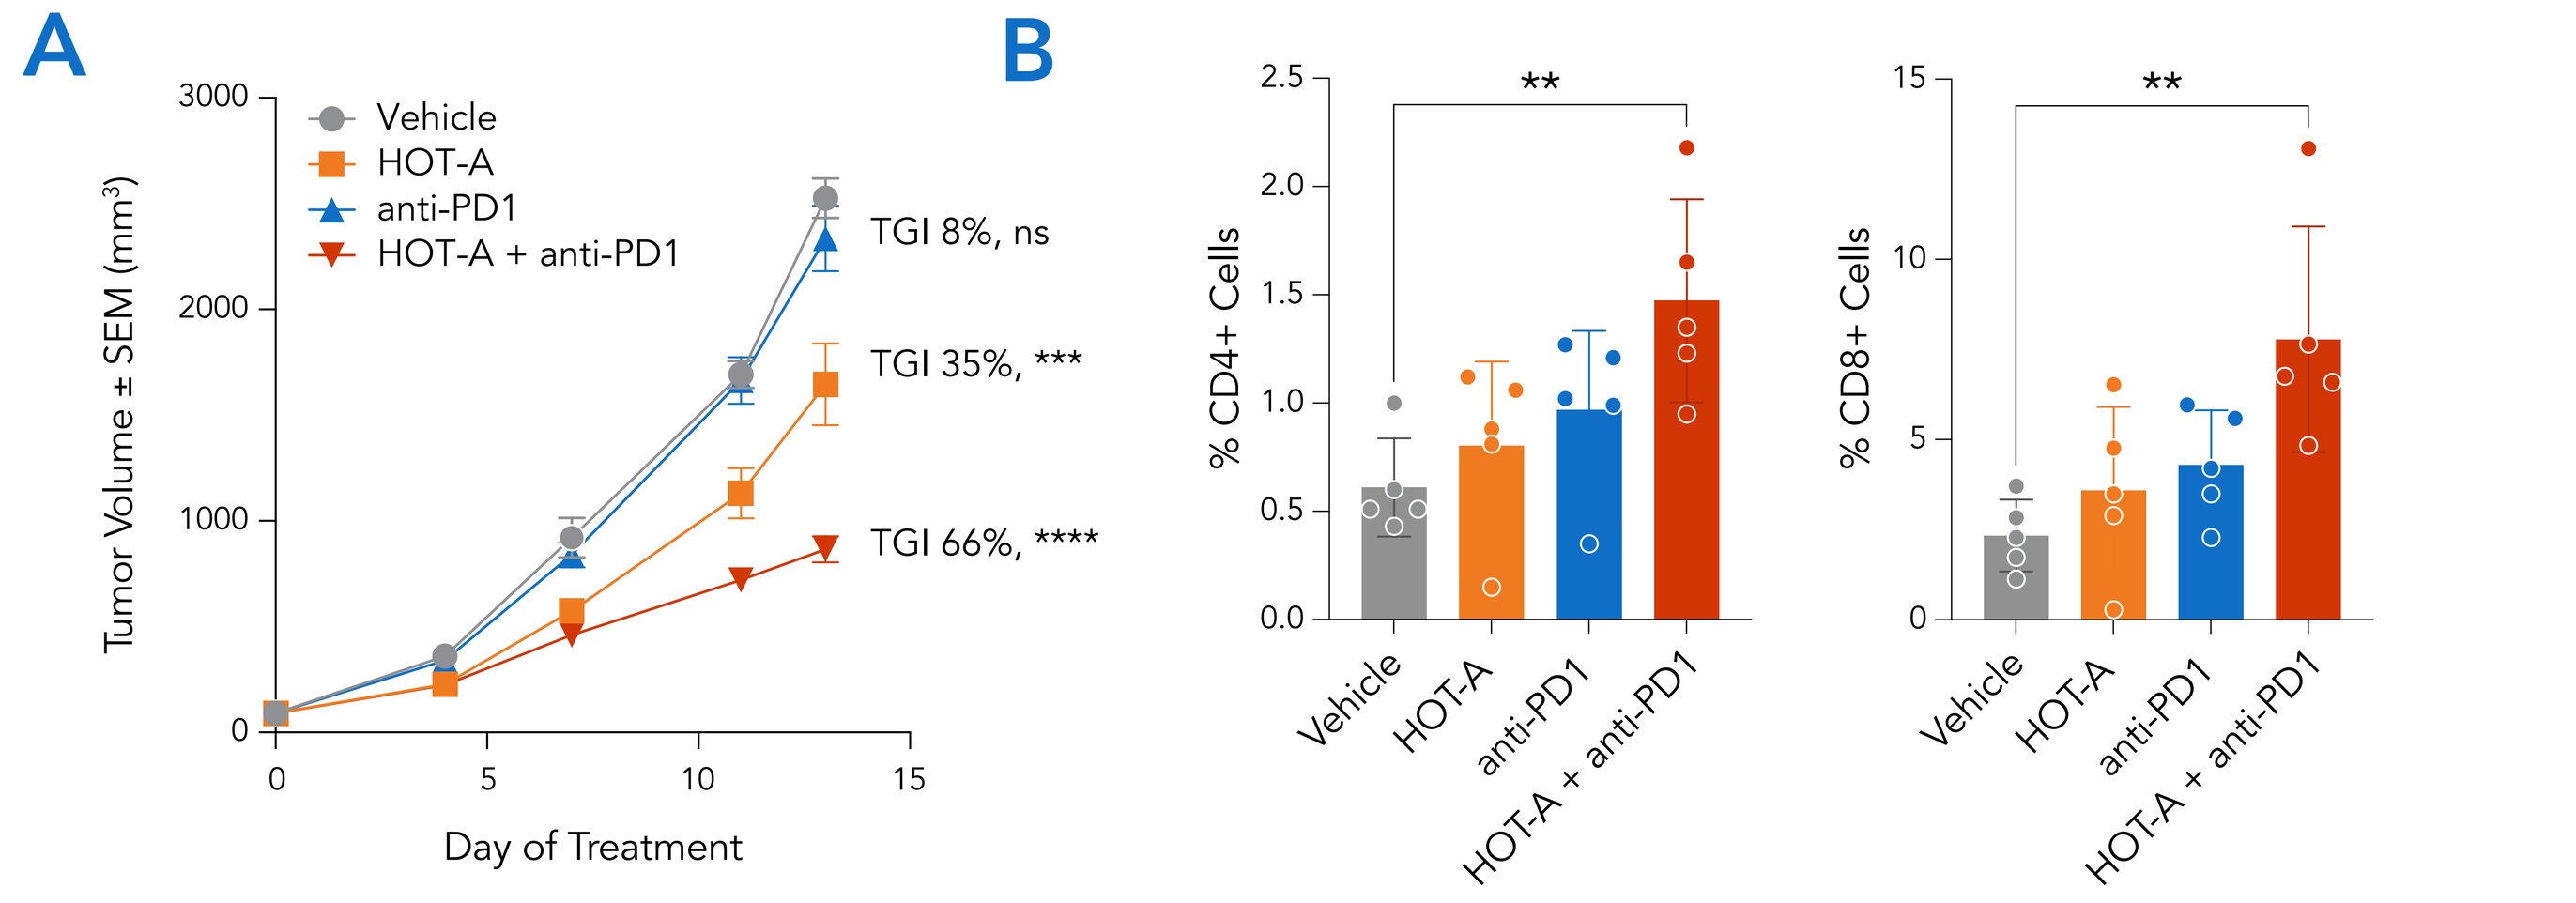
A
Vehicle
HOT-A
anti-PD1
HOT-A + anti-PD1
TGI 8%, ns
TGI 35%, ***
TGI 66%, ****
Tumor Volume ± SEM (mm³)
Day of Treatment
B
% CD4+ Cells
% CD8+ Cells
**
Vehicle
HOT-A
anti-PD1
HOT-A + anti-PD1

Female BALB/c mice were inoculated with CT26 tumor cells, randomized, and treated with vehicle, HOT-A, anti-PD1, or combination of HOT-A and anti-PD1. (A) Tumor growth curves for vehicle group and treated animals are shown. Data are displayed as mean ± standard error the mean (SEM). Statistics were calculated using one-way ANOVA on day 13 vehicle versus treated groups. ***$p<0.0001$; ****$p<0.00001$. (B) CD4+% and CD8+% cells in the tumors at day 10 by immunohistochemistry.

Figure 6. Combination of HOT-A and Anti-PD1 Promoted T Cell Infiltration and Reduced M2 Type Tumor Associated Macrophages

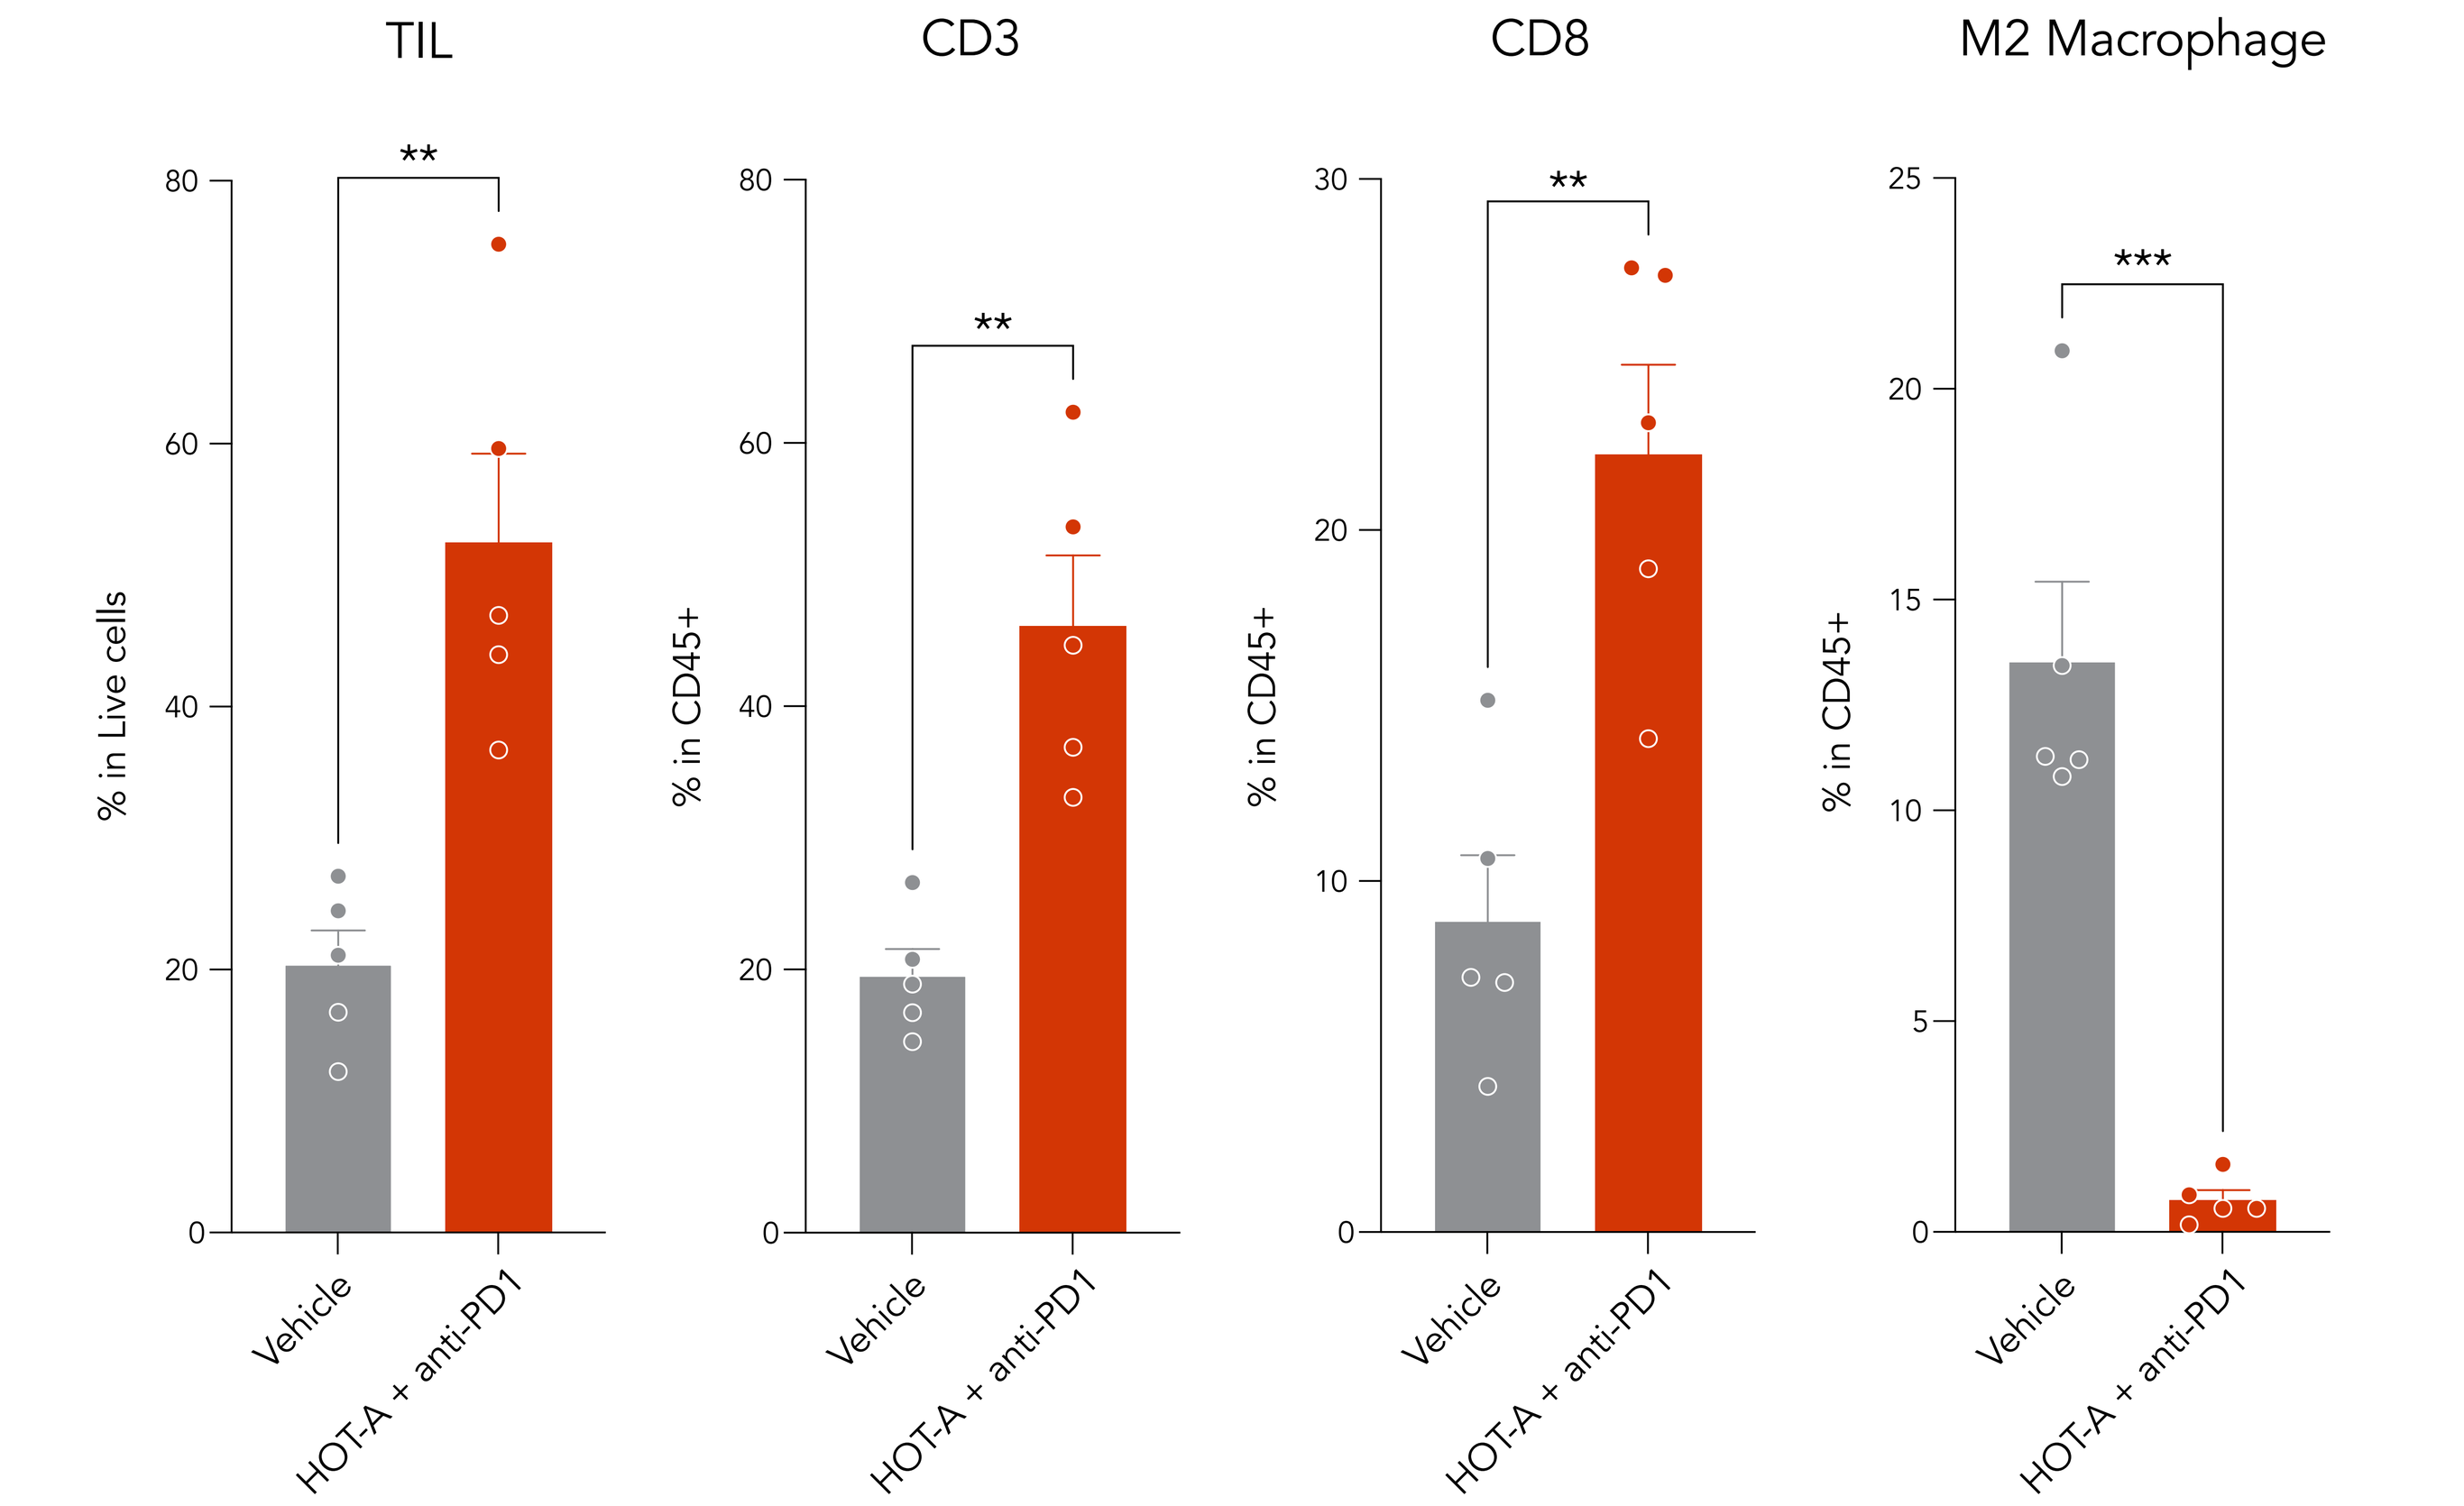
TIL
CD3
CD8
M2 Macrophage
% in Live cells
% in CD45+
**

Vehicle
HOT-A + anti-PD1

Female BALB/c mice were inoculated with CT26 tumor cells, randomized, and treated with vehicle, HOT-A, anti-PD1, or combination of HOT-A and anti-PD1. The frequency of TIL (CD45⁺) in the tumors, and the frequency of CD3, CD8 T cells, and CD206⁺MHCII$^{low/-}$ (M2 macrophage) within CD45⁺ populations. Statistics were calculated using t test. Only statistically significant differences among untreated and treated groups are shown. **$p<0.01$; ***$p<0.001$.

Conclusions

- HotSpot's CBL-B inhibitor, HOT-A, inhibited tumor growth in multiple murine syngeneic tumor models, including H22, CT26 and B16F10. The tumor inhibition was immune-mediated.
- Profiling tumor microenvironment gene expression demonstrated T cell function pathway, interferon pathway, antigen processing pathway were among the significantly up-regulated immune signature with HOT-A treatment *in vivo*.
- CBL-B inhibitor and anti-PD1 combination further enhanced tumor inhibition in selected tumor models. HOT-A and anti-PD1 combination further promoted T cell infiltration and shifted immune suppressive M2 macrophage to immune promoting M1 macrophage.
- Preclinical characterization both *in vitro* and *in vivo* suggests the monotherapy potential of this CBL-B inhibitor as well as the potential as a combinational agent with anti-PD1 antibody.

References

1. Lutz-Nicoladoni C, Wolf D, Sopper S. *Front Oncol.* 2015;5:58.
2. Bachmaier K, Krawczyk C, Kozieradzki I, et al. *Nature.* 2000;403(6766):211-216.
3. Chiang YJ, Kole HK, Brown K, et al. *Nature.* 2000;403(6766):216-220.
4. Wohlfert EA, Callahan MK, Clark RB. *J Immunol.* 2004;173(2):1059-1065.
5. Wohlfert EA, Gorelik L, Mittler R, Flavell RA, Clark RB. *J Immunol.* 2006;176(3):1316-1320.
6. Loeser S, Loser K, Bijker MS, et al. *J Exp Med.* 2007;204(4):879-891.
7. Chiang JY, Jang IK, Hodes R, Gu H. *J Clin Invest.* 2007;117(4):1029-1036.
8. Paolino M, Choidas A, Wallner S, et al. *Nature.* 2014;507(7493):508-512.

HOTSPOT THERAPEUTICS HotSpot Therapeutics, Inc • 50 Milk St, 16th floor • Boston, MA 02109 • USA • hotspotthera.com

Contact: Natalie Wildenradt - nwildenradt@hotspotthera.com

SITC 37th Annual Meeting • Boston, MA • November 8-12, 2022