A Novel Allosteric CBL-B Inhibitor with Differentiated Immune Enhancing Activity in Preclinical Models

#598

Yilin Qi*, Jun Kuai*, Yingzhi Bi*, Huadong Sun, Deborah G. Conrady, Rajiv G. Govindaraj, Graham Hone, Rajiah A. Denny, Ken Carson,Timothy Reilly, Geraldine Harriman, Fang Wang†

HotSpot Therapeutics, Inc, 50 Milk St, 16th floor, Boston, MA 02109, USA; *equal contribution; †corresponding author

Introduction

- E3 ligase Casitas B-Lineage Lymphoma Proto-Oncogene B (CBL-B) is a key negative modulator of T-cell receptor and co-stimulatory regulation.[1]
- CBL-B inhibition lowers the threshold of antigen-specific T cell activation, even in the absence of co-stimulatory signaling or the presence of an immune suppressive environment.[2-5]
- Genetic ablation of CBL-B or functional inactivation of its E3 ligase activity in mice or primary human T cells enhances immune-mediated tumor growth control.[6-8]
- CBL-B inhibition may address the suboptimal response to current immunotherapies due to low inflammation, no/low co-stimulation signal or a high immune suppressive environment.

CBL-B Inhibition Enhances Anti-tumor Immunity Through Several Key Biological Mechanisms

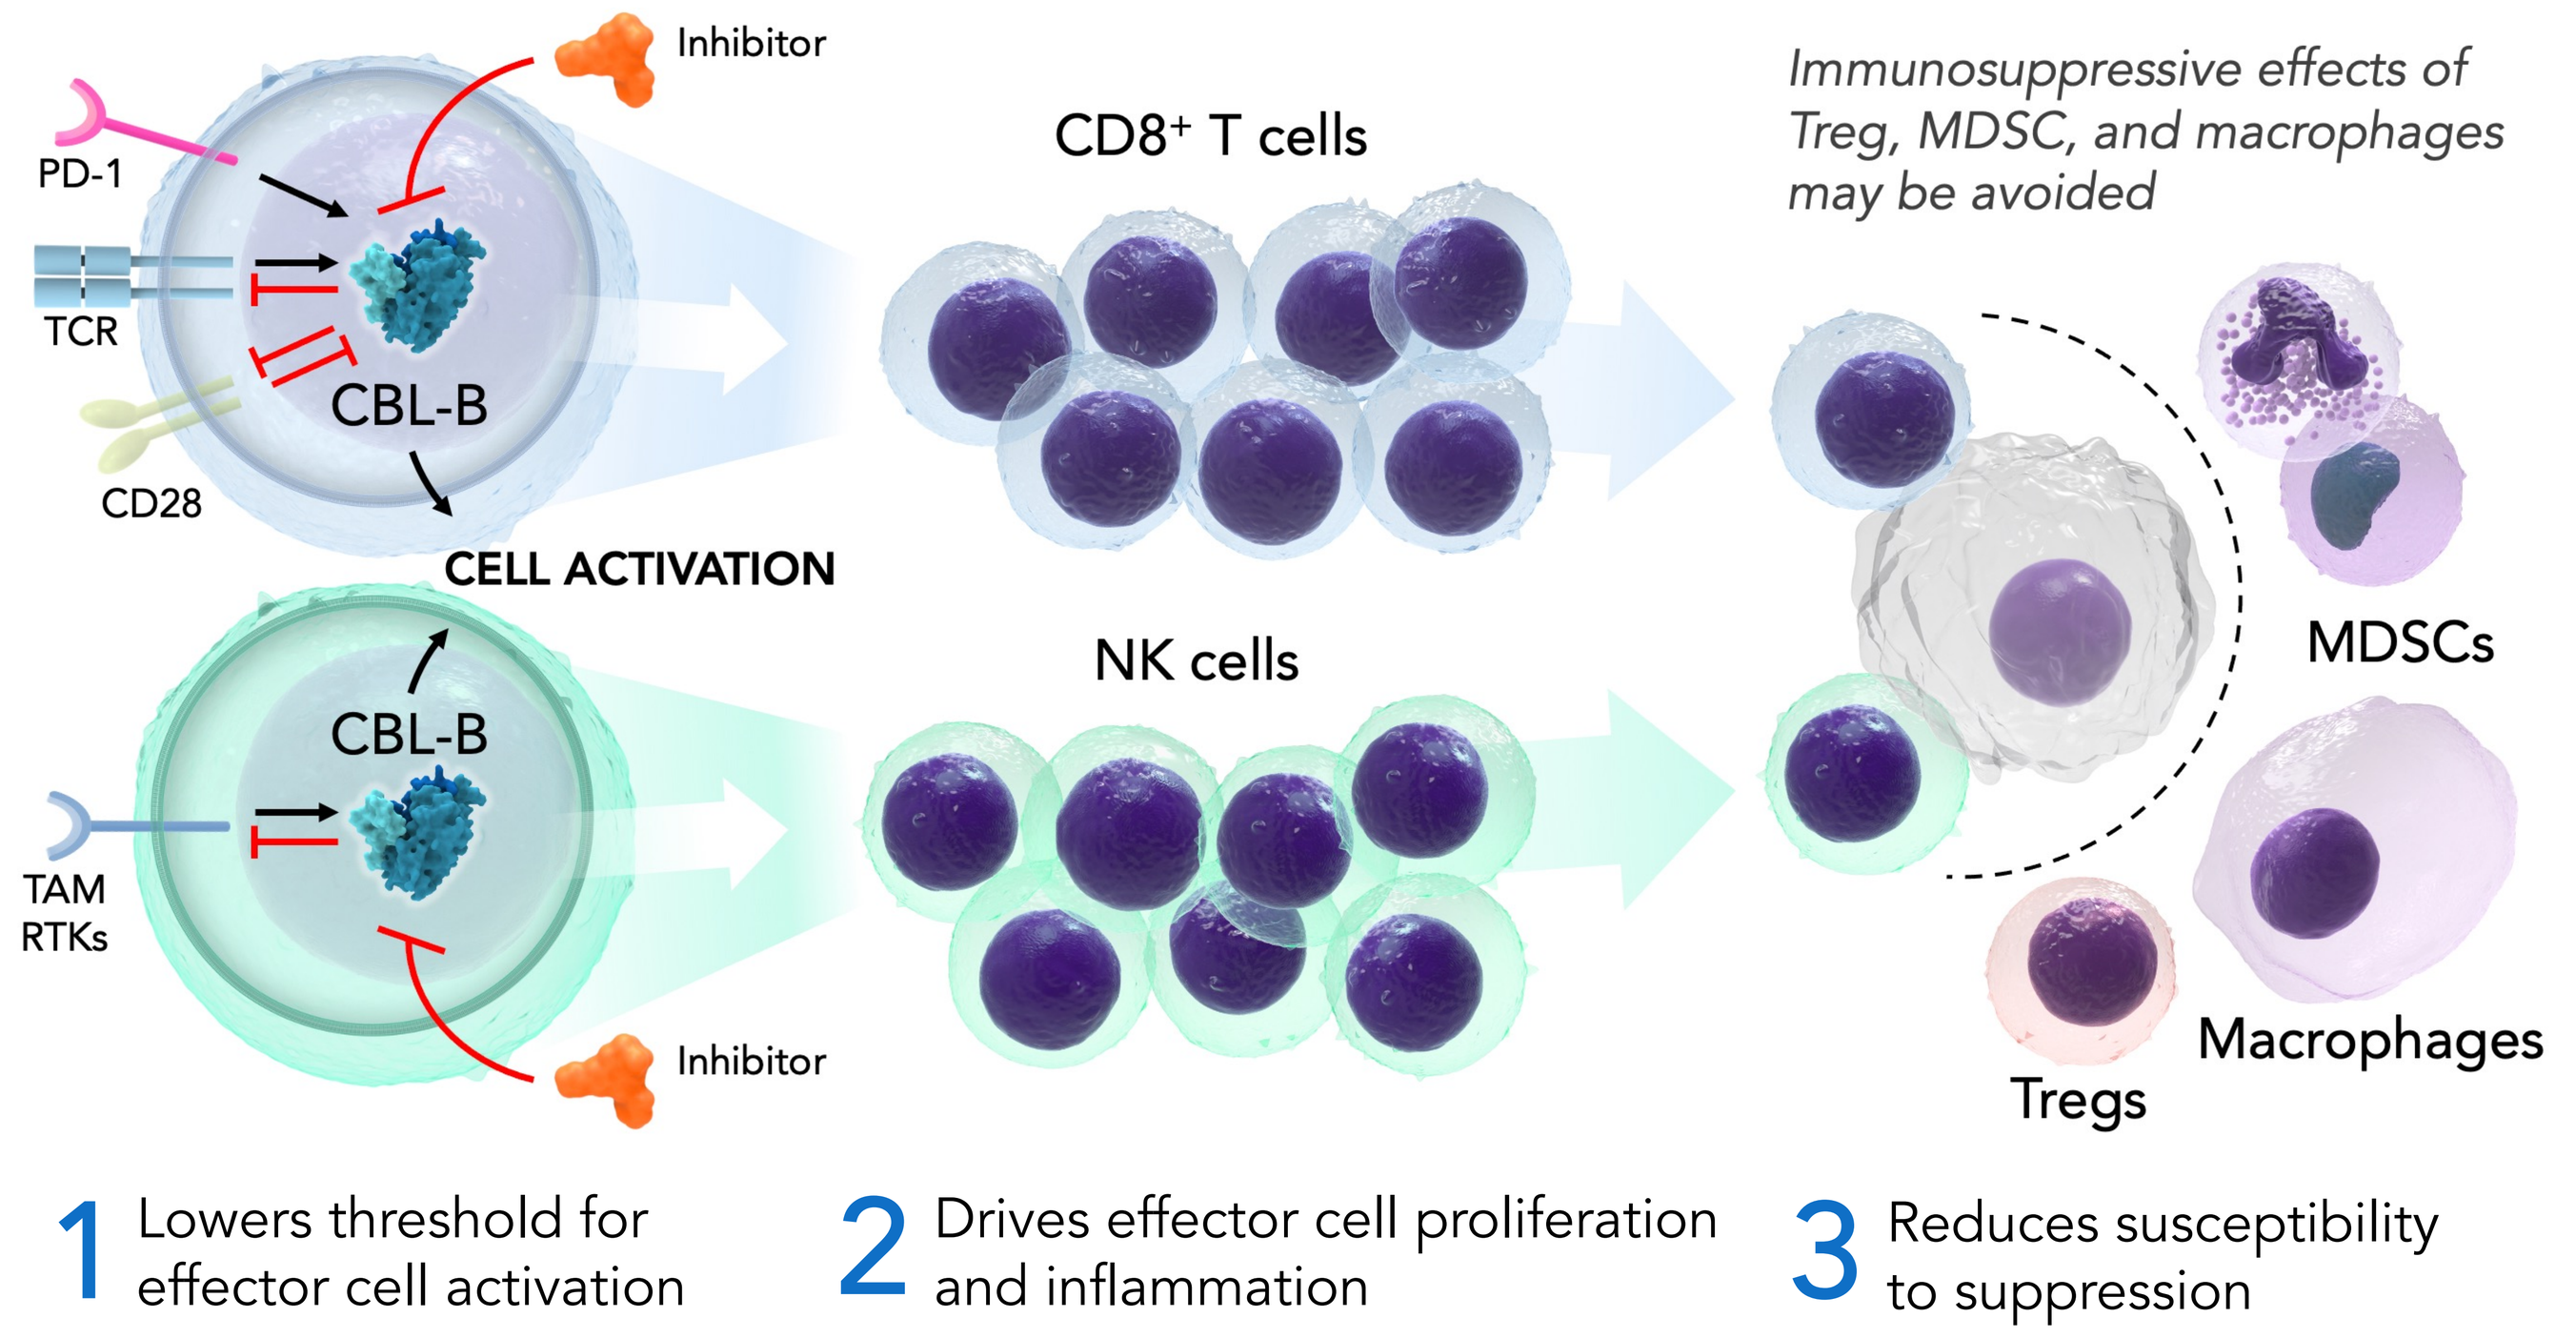
Inhibitor
PD-1
TCR
CBL-B
CD28
CELL ACTIVATION
CD8+ T cells
Immunosuppressive effects of Treg, MDSC, and macrophages may be avoided
NK cells
TAM RTKs
MDSCs
Macrophages
Tregs
1 Lowers threshold for effector cell activation
2 Drives effector cell proliferation and inflammation
3 Reduces susceptibility to suppression

Results

Table 1. HotSpot CBL-B Inhibitor, HOT-A, Shows Potent Immunostimulatory Activity *in vitro*

	Biochemical Assay	Human T Cell Assay			
Compound	TR-FRET CBL-B IC_{50} (nM)	CD4 proliferation EC_{50} (nM)	CD8 proliferation EC_{50} (nM)	IL-2 EC_{50} (nM)	IFNγ EC_{50} (nM)
HOT-A	6	0.8	0.6	18	30
REF	15	5	6	230	510

TR-FRET assay measures the interaction between the Biotin-E2 and CBL-B; Human T cell assay characterizes the effect of compounds on anti-CD3-stimulated human CD4+ and CD8+ T cells without anti-CD28 costimulation. IL-2 (24hr) and IFNγ (72hr) secretion were measured from supernatant by ELISA. The frequency of T cell proliferation at 72 hours was assessed through T cell labeled cell trace violet dye dilution by flow cytometry. REF: synthesized small molecule based on example 519 from patent WO 2019/148005 A1.[9]

Figure 1. HOT-A Enhanced IL-2 Release in the *in vivo* Anti-CD3 PD Model

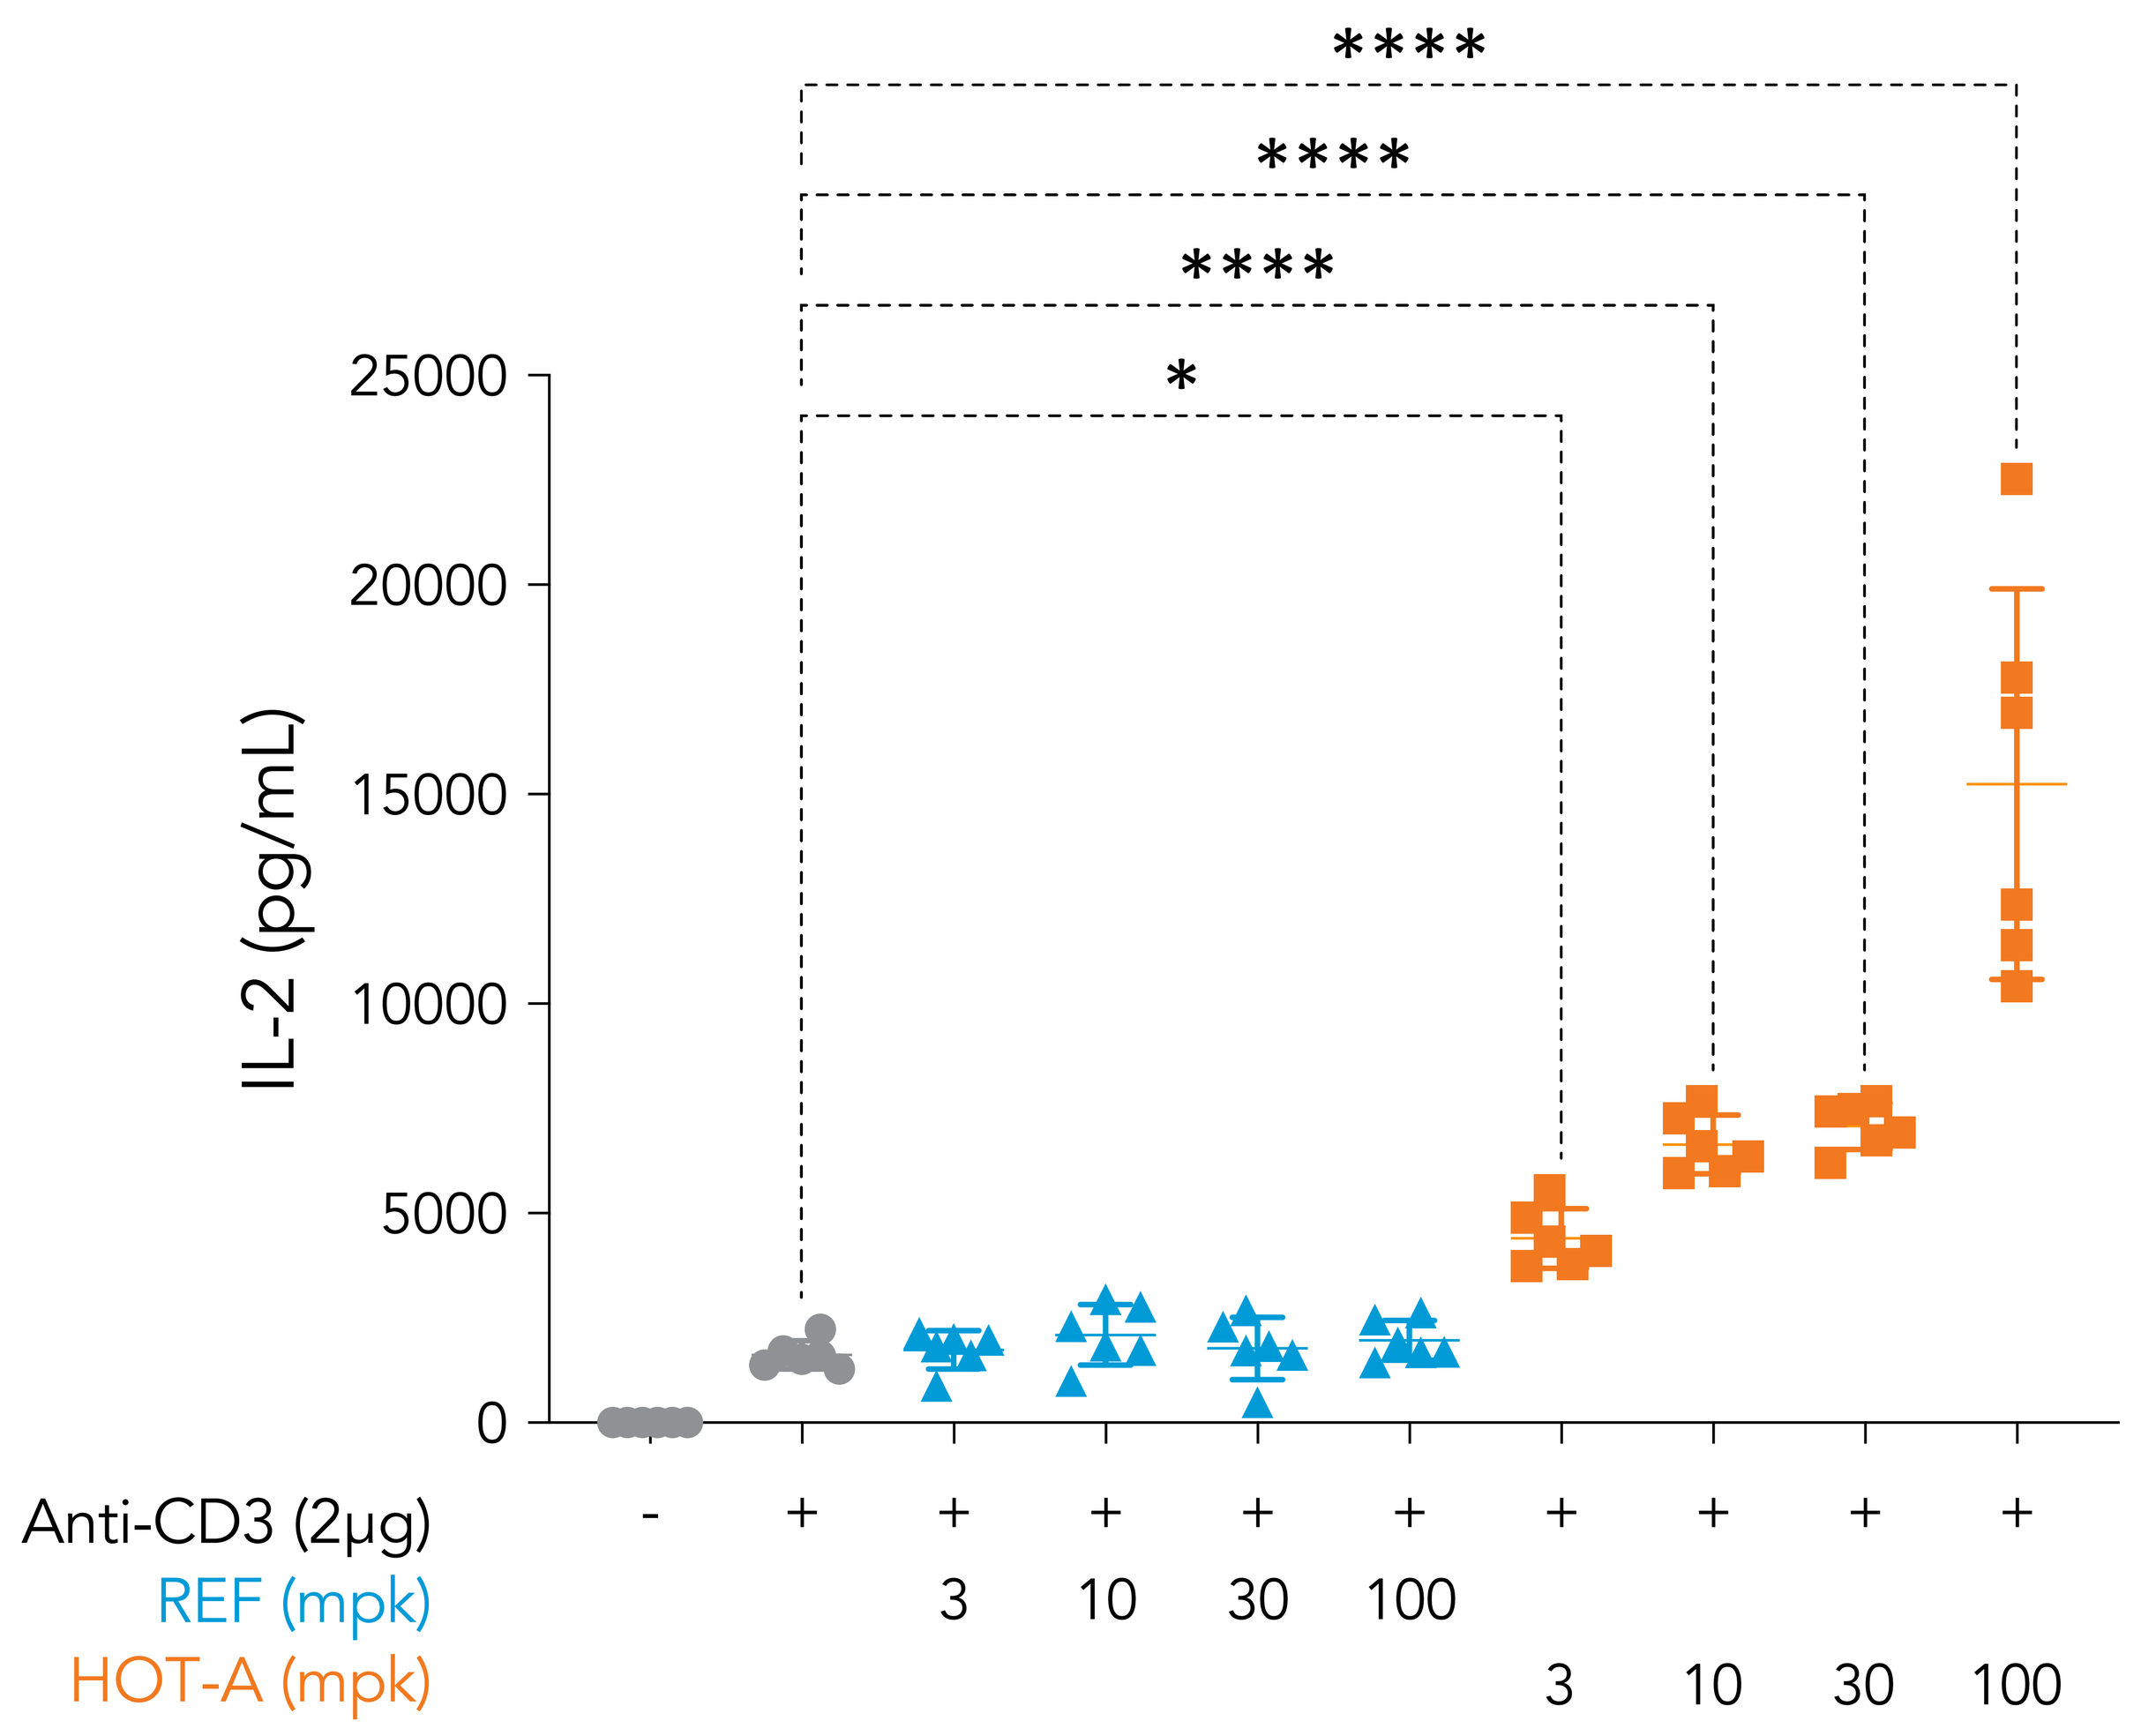
IL-2 (pg/mL)
25000
20000
15000
10000
5000
0

*
Anti-CD3 (2μg) - + + + + + + + + +
REF (mpk) 3 10 30 100
HOT-A (mpk) 3 10 30 100

Compound HOT-A or REF was orally (PO) administered to female BALB/c mice at indicated doses one hour before the intraperitoneal (IP) injection of anti-CD3 antibody. Serum IL-2 levels were measured 4 hours after the anti-CD3 injection by MSD.

Figure 2. HOT-A Showed Enhanced Tumor Growth Inhibition Versus Key Comparators in the CT26 Tumor Model

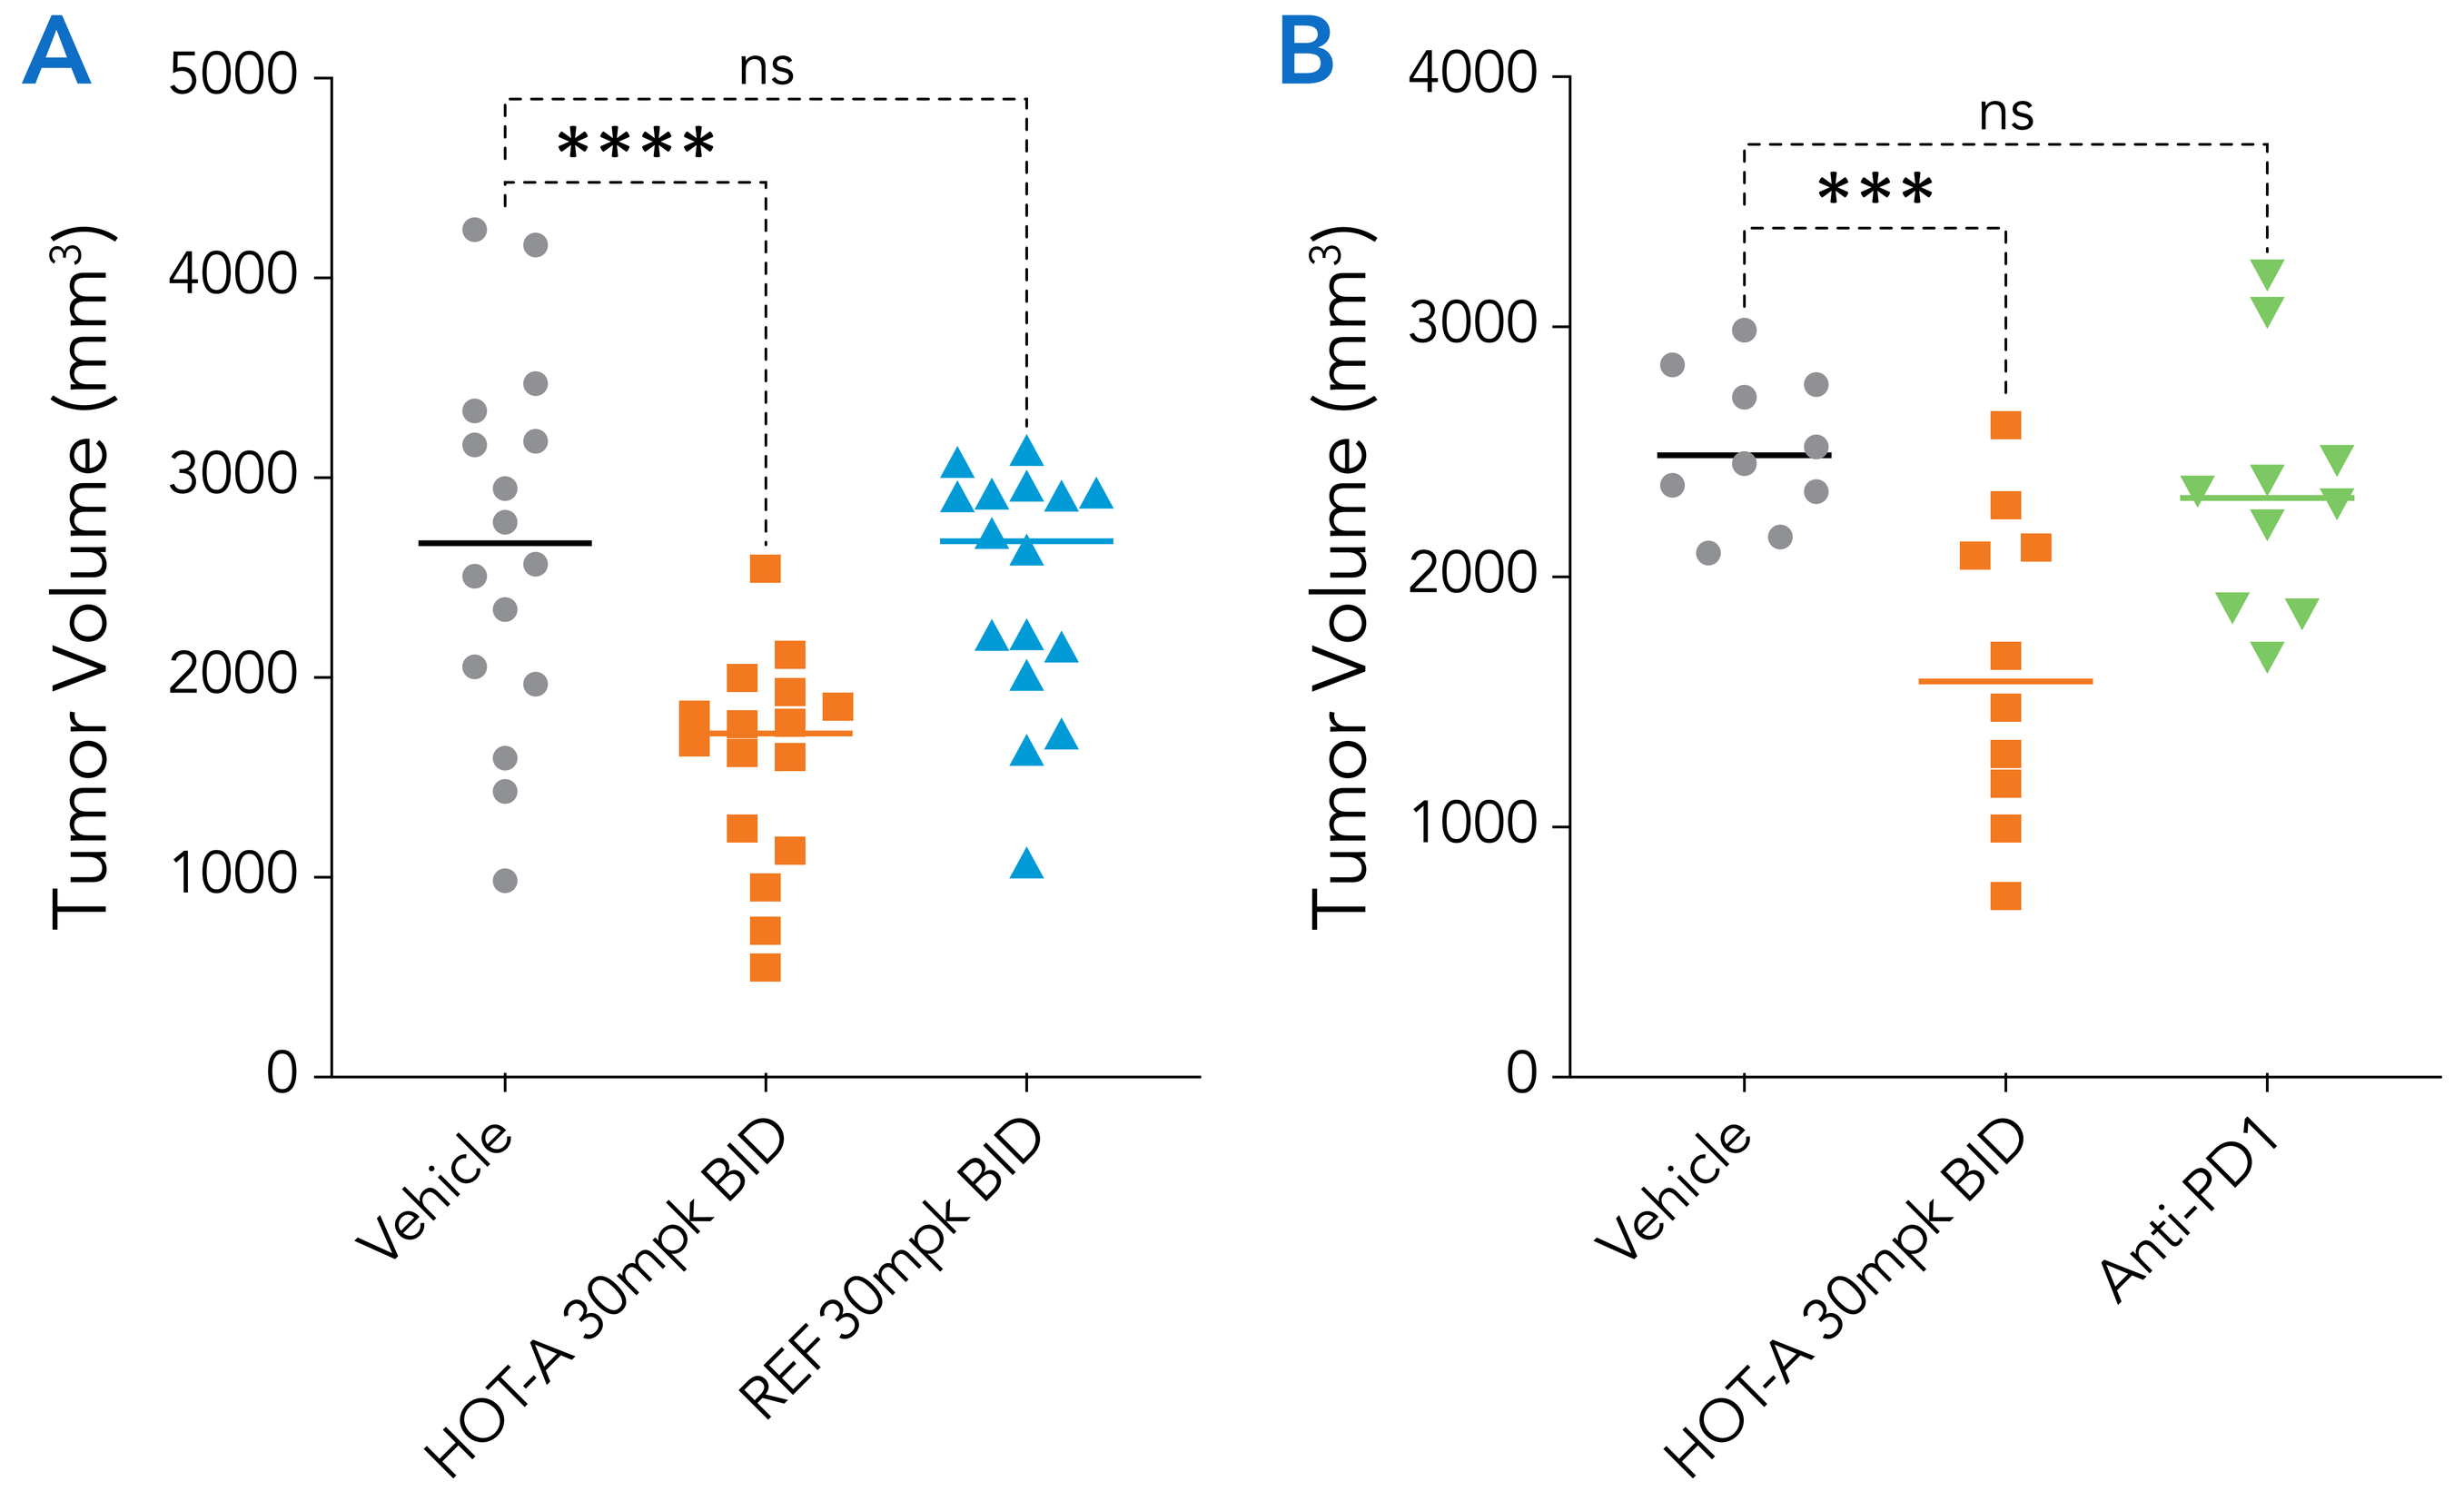
A
Tumor Volume (mm³)
5000
4000
3000
2000
1000
0
ns

Vehicle
HOT-A 30mpk BID
REF 30mpk BID
B
Tumor Volume (mm³)
4000
3000
2000
1000
0
ns

Vehicle
HOT-A 30mpk BID
Anti-PD1

Female BALB/c mice were inoculated with CT26 colorectal carcinoma cells, randomized, and treated with vehicle, HOT-A (30mpk, BID, PO), REF (30mpk, BID, PO), anti-PD1 (10mpk, BIW, IP) or isotype control. (A) Individual tumor volume on day 17 in vehicle, HOT-A or REF treated tumor-bearing mice. (B) Individual tumor volume on day 13 in vehicle + isotype control (Vehicle), HOT-A + isotype control (HOT-A 30mpk BID) or vehicle + anti-PD1 (Anti-PD1) treated tumor-bearing mice.

Figure 3. CBL-B Inhibition Restored IFNγ Secretion in the Exhausted T Cells (Tex)

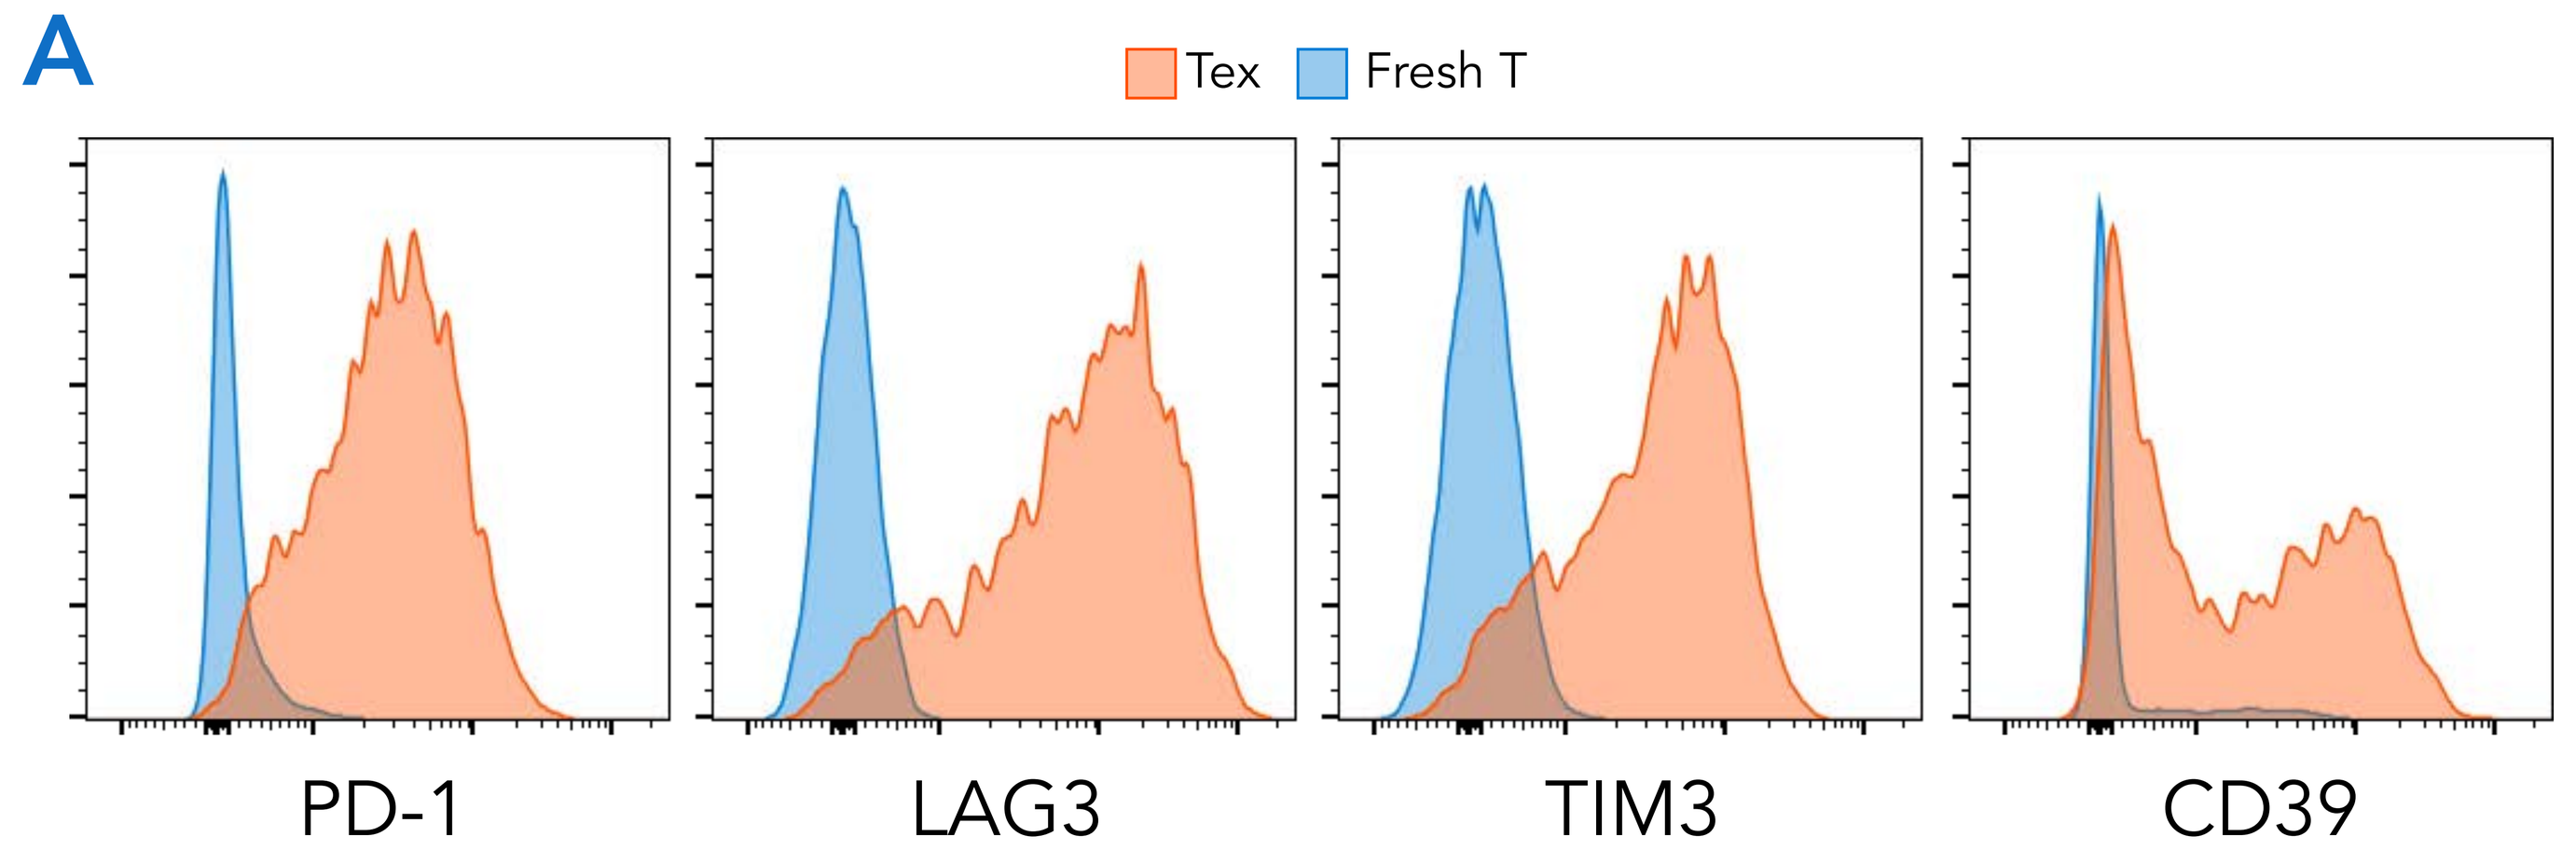
A
Tex
Fresh T
PD-1
LAG3
TIM3
CD39

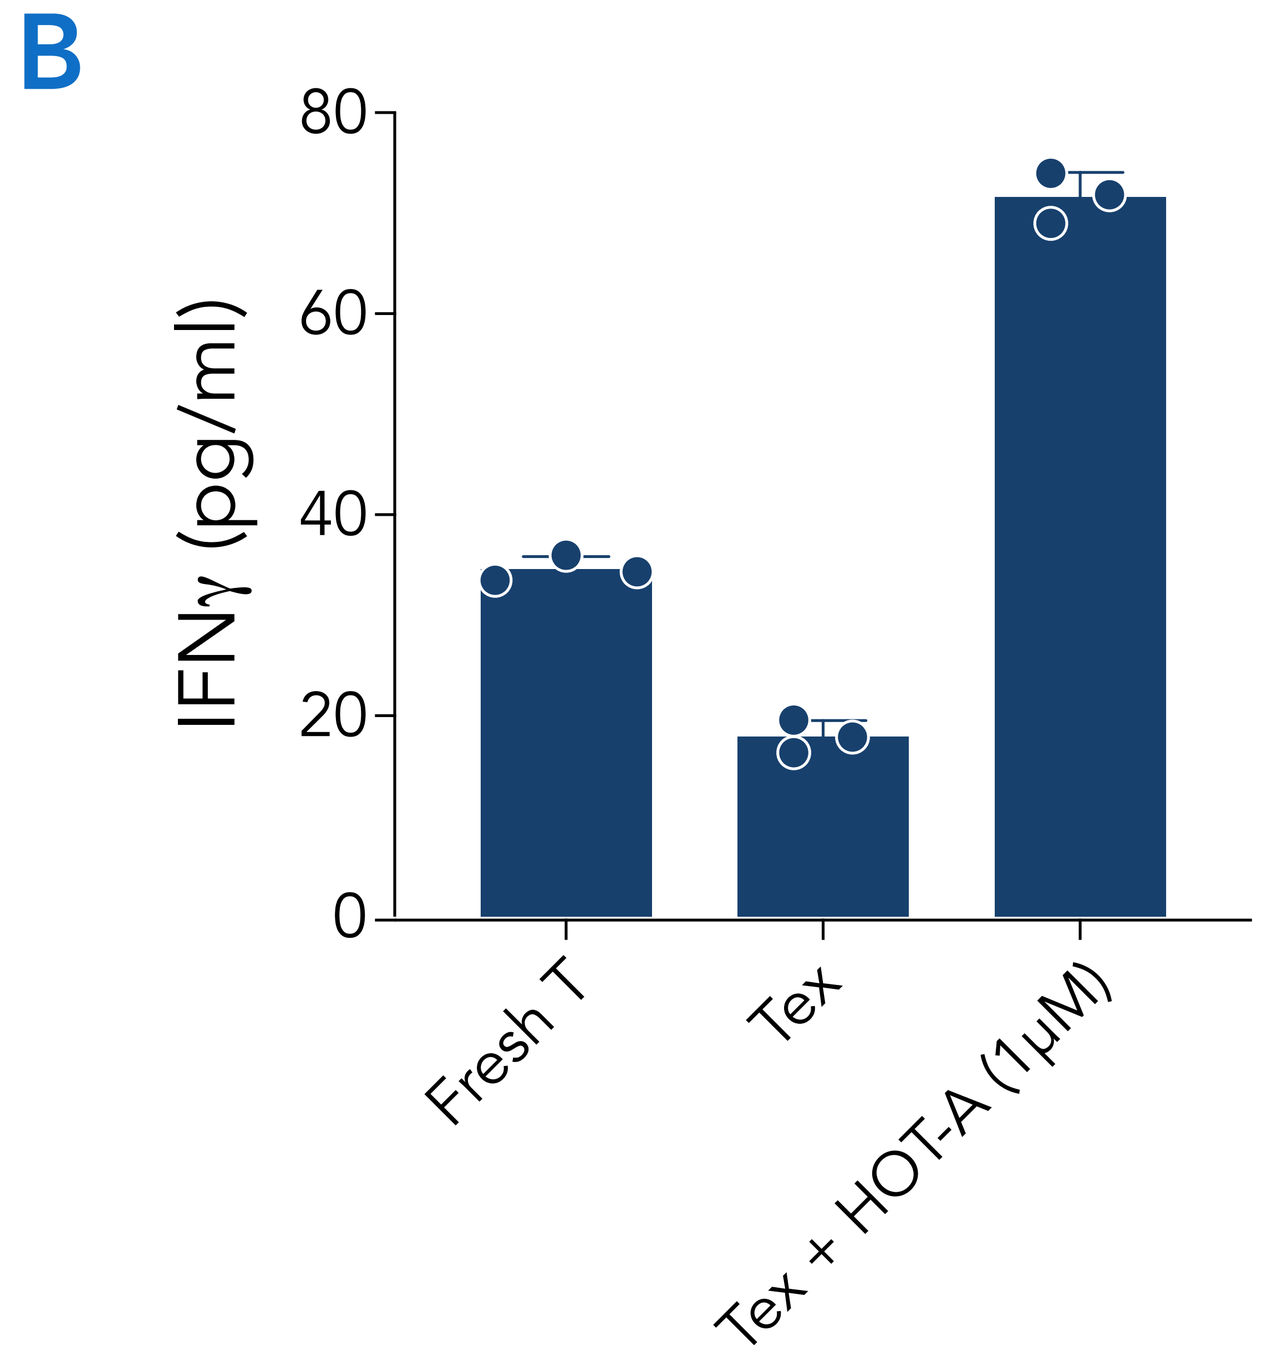
B
IFNγ (pg/ml)
80
60
40
20
0
Fresh T
Tex
Tex + HOT-A (1μM)

Tex were induced by repetitive *in vitro* anti-CD3/CD28 stimulation. (A) Tex (orange) showed increased surface PD1, LAG3, TIM3 and CD39 expression compared to fresh isolated T cells (Fresh T, blue). (B) Tex and Fresh T were stimulated with anti-CD3/CD28 with or without HOT-A (1μM). IFNγ secretion were analyzed after 72 hour treatment.

Figure 4. CBL-B Inhibition Restored the Teff Response to Treg Suppression

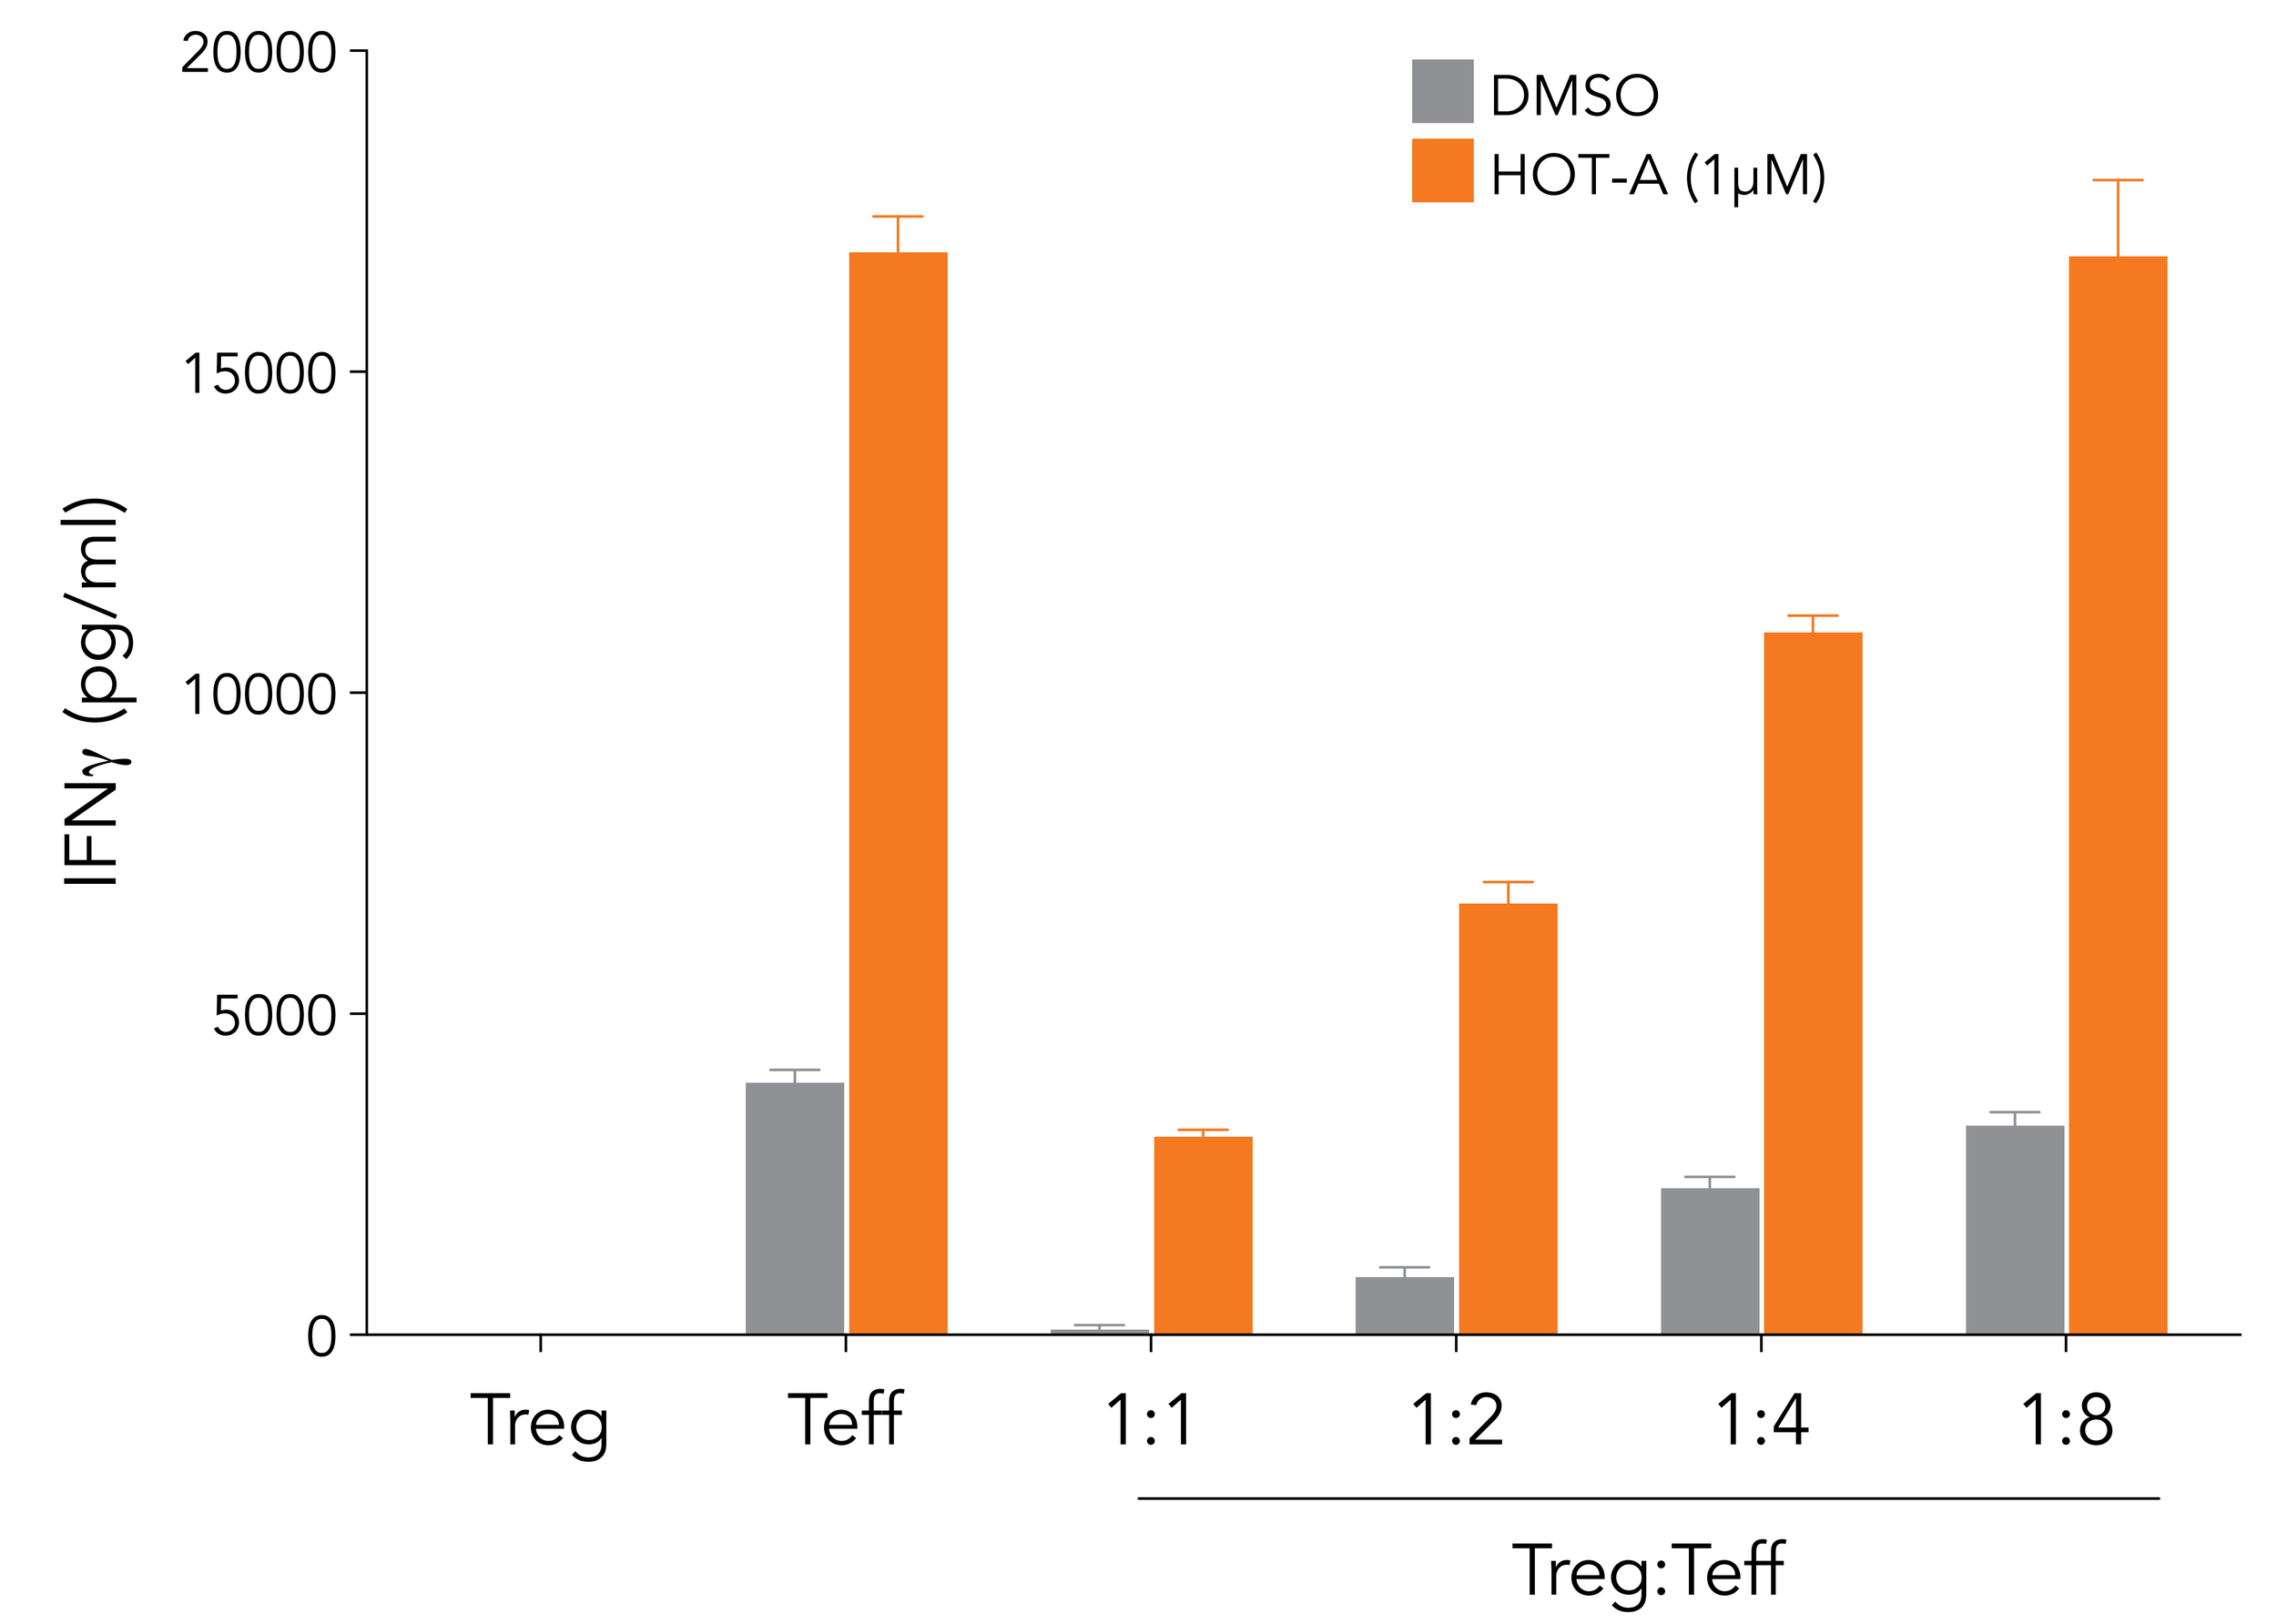
IFNγ (pg/ml)
20000
15000
10000
5000
0
DMSO
HOT-A (1μM)
Treg
Teff
1:1
1:2
1:4
1:8
Treg:Teff

Human CD4+CD127lowCD25+ regulatory T cells were isolated from human PBMC. After *in vitro* expansion for 14 days, coculture of Treg and Teff at various ratios were set up in the presence of anti-CD3/CD28 stimulation and HOT-A (1μM). IFNγ secretion was measured at 72 hours after the coculture.

Figure 5. CBL-B Inhibition Enhances MLR Responses as Monotherapy with Further Enhancement in Combination with Anti-PD1

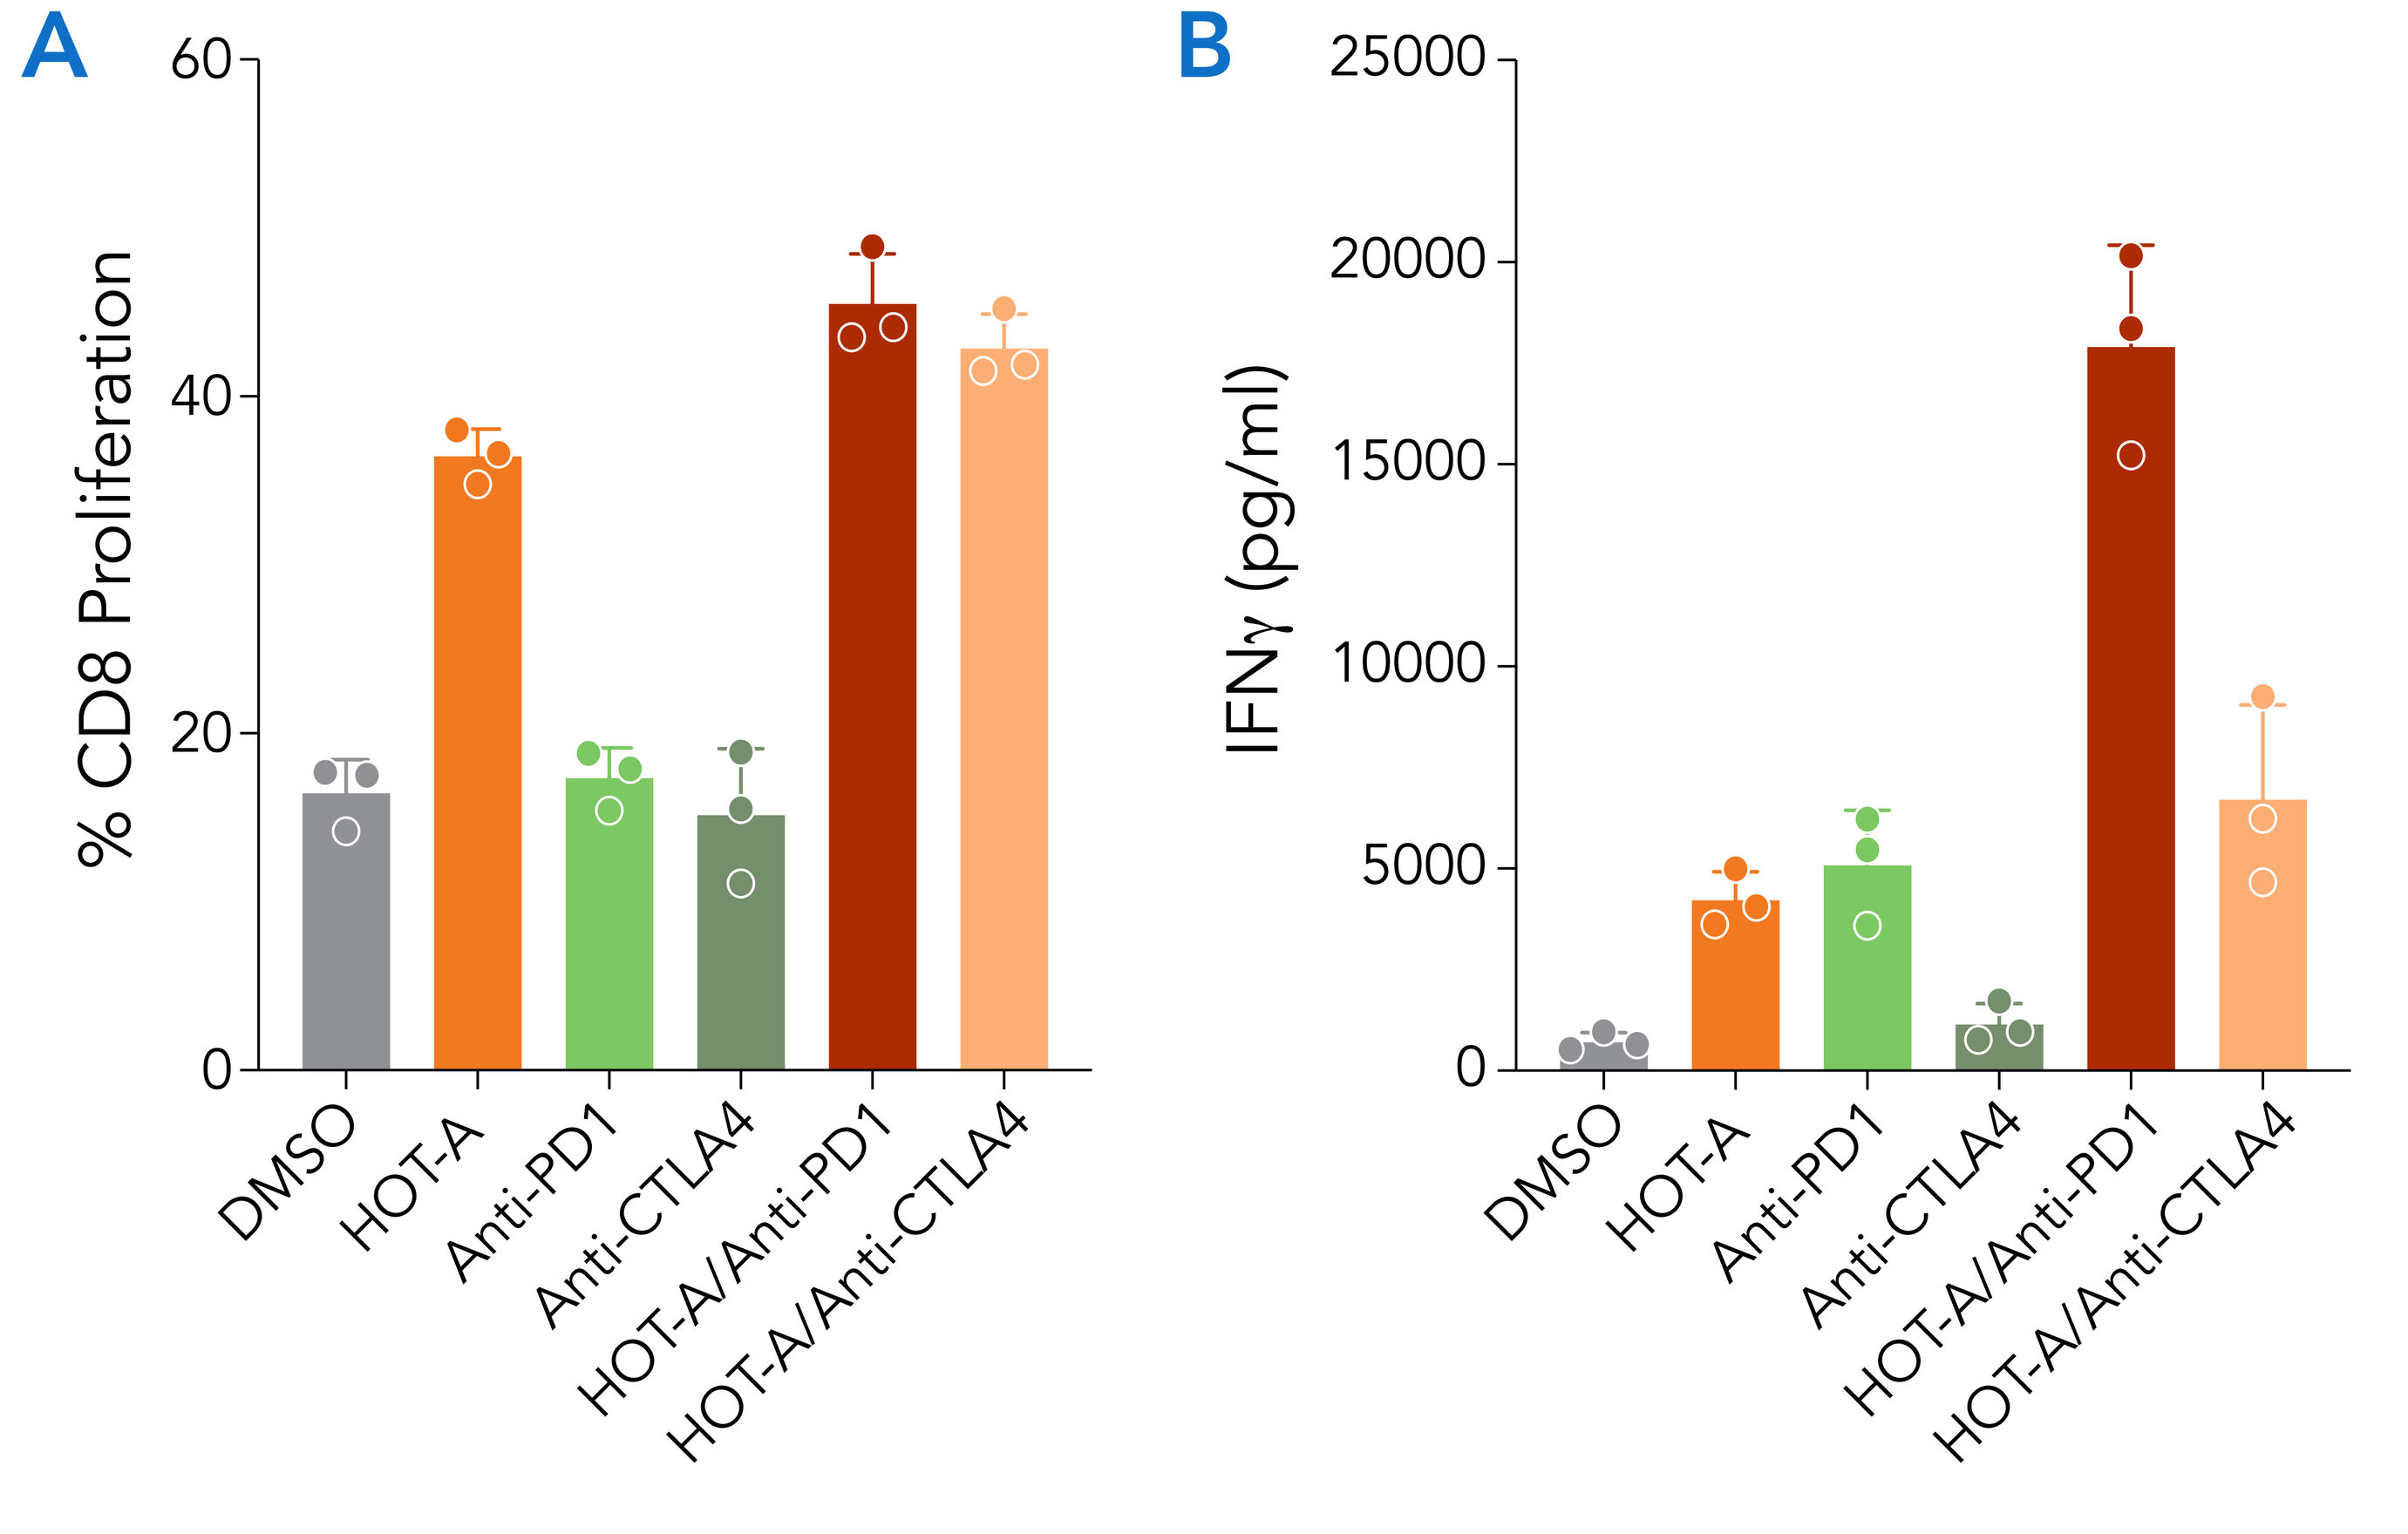
A
% CD8 Proliferation
60
40
20
0
DMSO
HOT-A
Anti-PD1
Anti-CTLA4
HOT-A/Anti-PD1
HOT-A/Anti-CTLA4
B
IFNγ (pg/ml)
25000
20000
15000
10000
5000
0
DMSO
HOT-A
Anti-PD1
Anti-CTLA4
HOT-A/Anti-PD1
HOT-A/Anti-CTLA4

T cells from donor one PBMC were isolated and labeled with CellTraceViolet (CTV). Labeled T cells were then mixed and cultured with dendrite cells from a different donor (allo DC). After 4 day coculture in the presence or absence of HOT-A (1μM), anti-PD1 (10μg/ml) or anti-CTLA4 (10μg/ml), (A) proliferation was measured by the frequency of T cells with decreased CTV fluorescence and (B) IFNγ secretion was assessed by ELISA.

Conclusions

- We have identified HOT-A, a novel allosteric small molecule that potently inhibits CBL-B E3 ubiquitin ligase activity.
- HOT-A demonstrated superior activity *in vitro* and *in vivo* compared to key comparators.
- HOT-A showed an ability to reinvigorate exhaustive T cells. Additionally, treatment with HOT-A restored the Teff response in the presence of Treg-mediated suppression.
- HOT-A synergizes with anti-PD1 to enhance T cell proliferation and IFNγ in MLR assay.
- Our allosteric CBL-B inhibitors enhance key parameters associated with T cell activation and reduce susceptibility to immune suppression, supporting their further progression toward clinical development.

References

1. Lutz-Nicoladoni C, Wolf D, Sopper S. *Front Oncol.* 2015;5:58.
2. Bachmaier K, Krawczyk C, Kozieradzki I, et al. *Nature.* 2000;403(6766):211-216.
3. Chiang YJ, Kole HK, Brown K, et al. *Nature.* 2000;403(6766):216-220.
4. Wohlfert EA, Callahan MK, Clark RB. *J Immunol.* 2004;173(2):1059-1065.
5. Wohlfert EA, Gorelik L, Mittler R, Flavell RA, Clark RB. *J Immunol.* 2006;176(3):1316-1320.
6. Loeser S, Loser K, Bijker MS, et al. *J Exp Med.* 2007;204(4):879-891.
7. Chiang JY, Jang IK, Hodes R, Gu H. *J Clin Invest.* 2007;117(4):1029-1036.
8. Paolino M, Choidas A, Wallner S, et al. *Nature.* 2014;507(7493):508-512.
9. Example 519 from patent WO 2019/148005 A1.

HOTSPOT THERAPEUTICS
HotSpot Therapeutics, Inc • 50 Milk St, 16th floor • Boston, MA 02109 • USA • hotspotthera.com
Contact: Natalie Wildenradt - nwildenradt@hotspotthera.com
AACR Annual Meeting • New Orleans, Louisiana • April 8-13, 2022